Spatial transcriptomics analysis reveals association between DDR1-collagen interactions and immune exclusion in human cancers

James Rouse[1], Laura A. Dillon[1], Xinwei Sher[1], Amy Mueller[1]

[1] Incendia Therapeutics, Boston, MA, USA

INCENDIA THERAPEUTICS #173

Background

Discoidin domain receptor 1 (DDR1) is overexpressed in many cancers and is a potential therapeutic target. DDR1 is implicated in immune exclusion and collagen alignment in *in vivo* models [1] and is associated with immune exclusion across human tumors based on analysis of H&E and multiplex immunofluorescence images, and bulk gene expression [2]. The emergence of spatial transcriptomics technology has enabled highly-multiplexed or whole-transcriptome spatially-resolved biomarker analysis. Here, we compiled and harmonized Visium spatial gene expression data (10x Genomics) from 224 human tumors across 14 tumor types and investigated the relationships between DDR1, DDR1-collagen co-expression, and immune exclusion.

Methods

Spatial gene expression data from human tumors were collected from various publications [3-21]. Gene expression was normalized to mean of 30,000 counts per spot per sample and using SCTransform within each sample to allow for comparison of gene expression across and within samples. Spots were clustered using Louvain clustering. Clusters were assigned as stromal or epithelial based on enrichment of a stromal gene signature [22]. The degree of CD8 exclusion was measured using the log2 ratio of average expression of a CD8 signature [23] in stromal vs. epithelial spots for each sample (Figure 1). Samples with mean CD8 signature scores in the lowest 20th percentile were removed prior to subsequent analysis. Ligand-receptor (LR) interaction analysis was performed using COMMOT [24]. LR co-expression is defined as the sum of signaling received across spots for each sample and for each LR pair from CellChatDB, CellTalkDB, and custom-defined DDR1-collagen interactions (Figure 2).

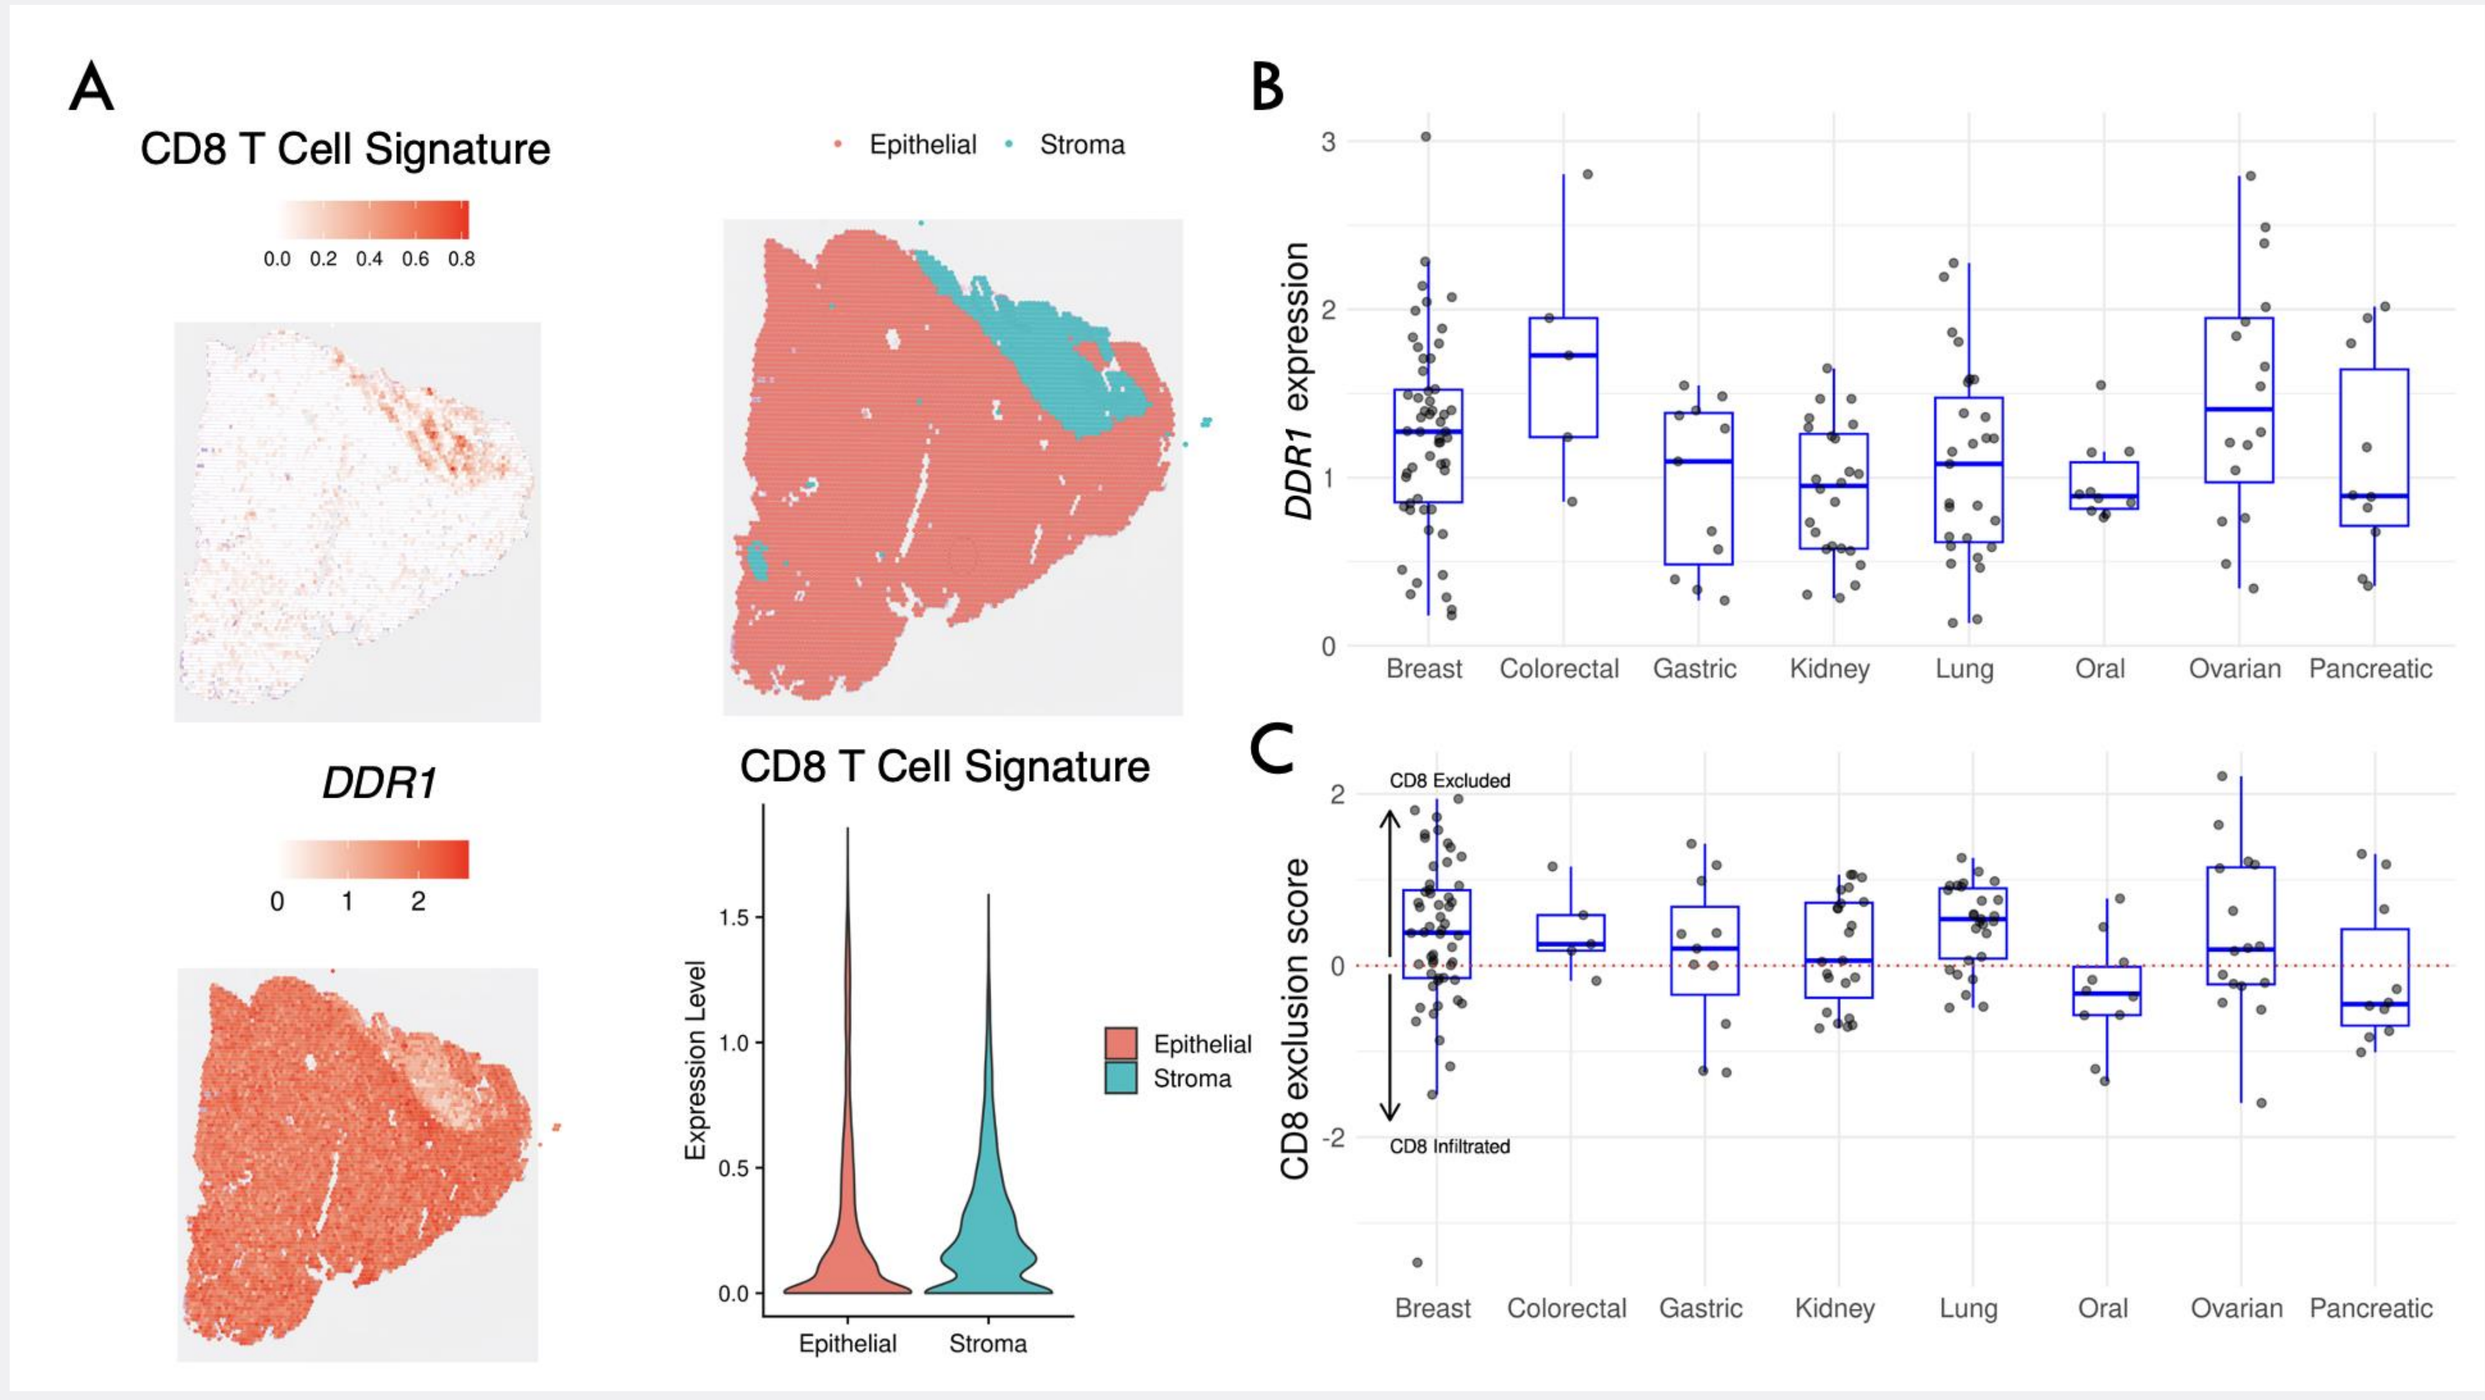
A
CD8 T Cell Signature
Epithelial
Stroma
DDR1
B
DDR1 expression
C
CD8 exclusion score
CD8 Excluded
CD8 Infiltrated
Breast
Colorectal
Gastric
Kidney
Lung
Oral
Ovarian
Pancreatic
Expression Level

Figure 1. Spatial quantification plots of CD8 signature expression and *DDR1* expression in a lung neuroendocrine tumor. A) CD8 signature expression is higher in the stroma in this sample, indicating that this sample is immune excluded. B) *DDR1* expression quantified across indications. C.) CD8 exclusion score quantified across indications.

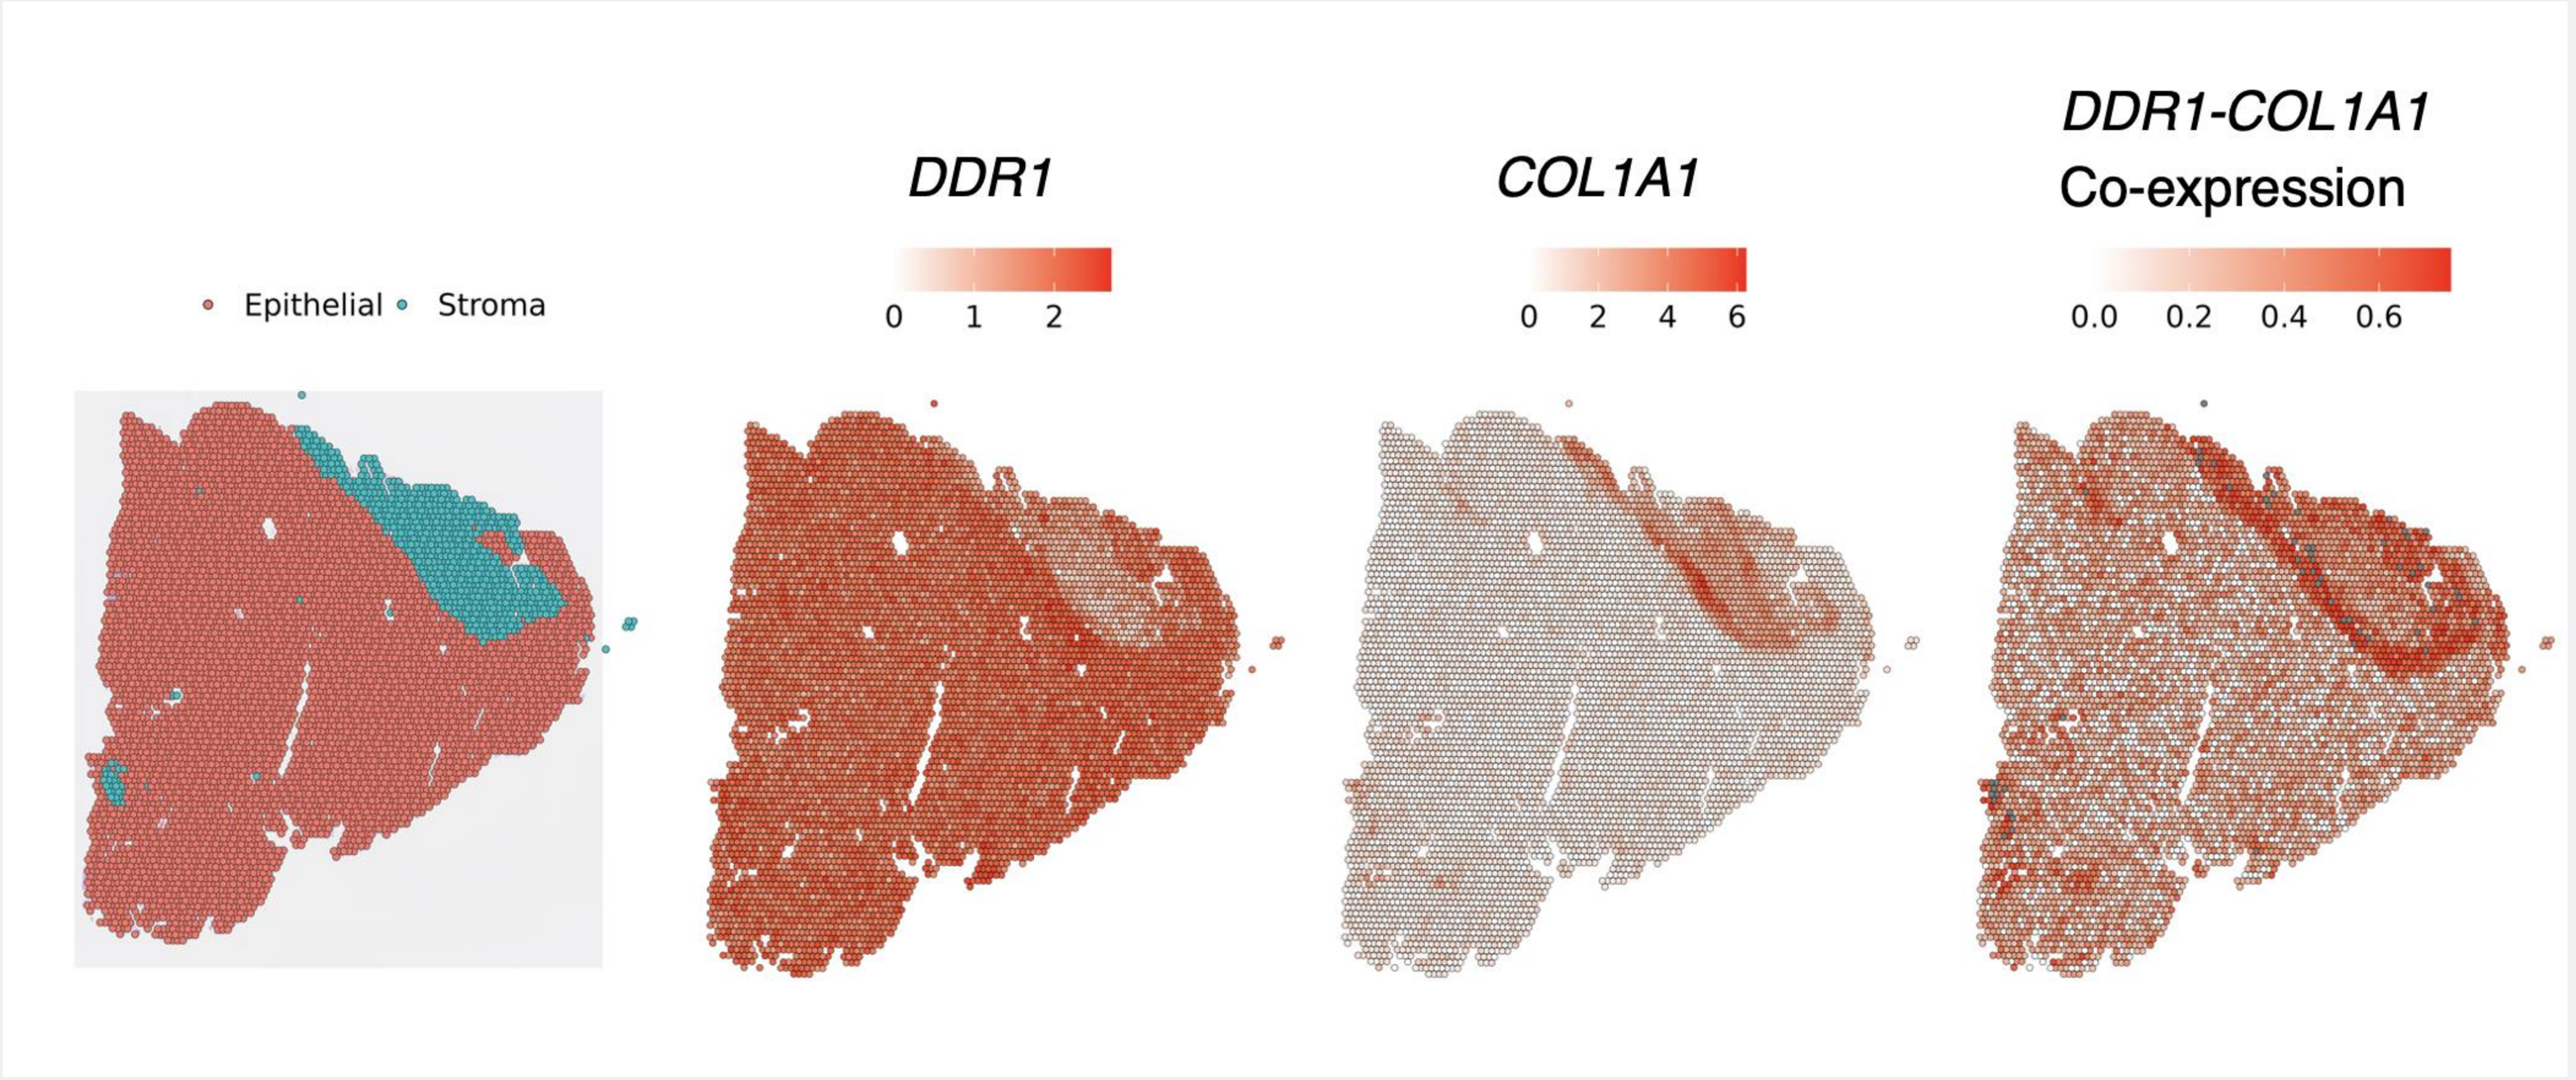
Epithelial
Stroma
DDR1
COL1A1
DDR1-COL1A1
Co-expression

Figure 2: Spatial analysis of stroma-epithelium distribution, *DDR1* and *COL1A1* expression, and *DDR1-COL1A1* co-expression in a lung neuroendocrine tumor sample. *DDR1-COL1A1* co-expression is highest at the stroma-epithelium interface.

Results

- High *DDR1* expression is associated with CD8 exclusion across the full dataset of 14 tumor types (r=0.27, p = 0.0006), with strongest association in hormone receptor positive breast cancer (r = 0.67, p = 0.0003) and non-small cell lung cancer (r = 0.58, p = 0.007) (Figure 3).
- *DDR1*-collagen interactions are significantly associated with CD8 exclusion for most collagen types evaluated (I, II, III, IV, V, VIII, XI) across tumor types and within tumor types including breast and gastric cancers (Figure 4).
- *DDR1*-collagen interactions are enriched among the top LR pathways associated with immune exclusion. (Figure 5)

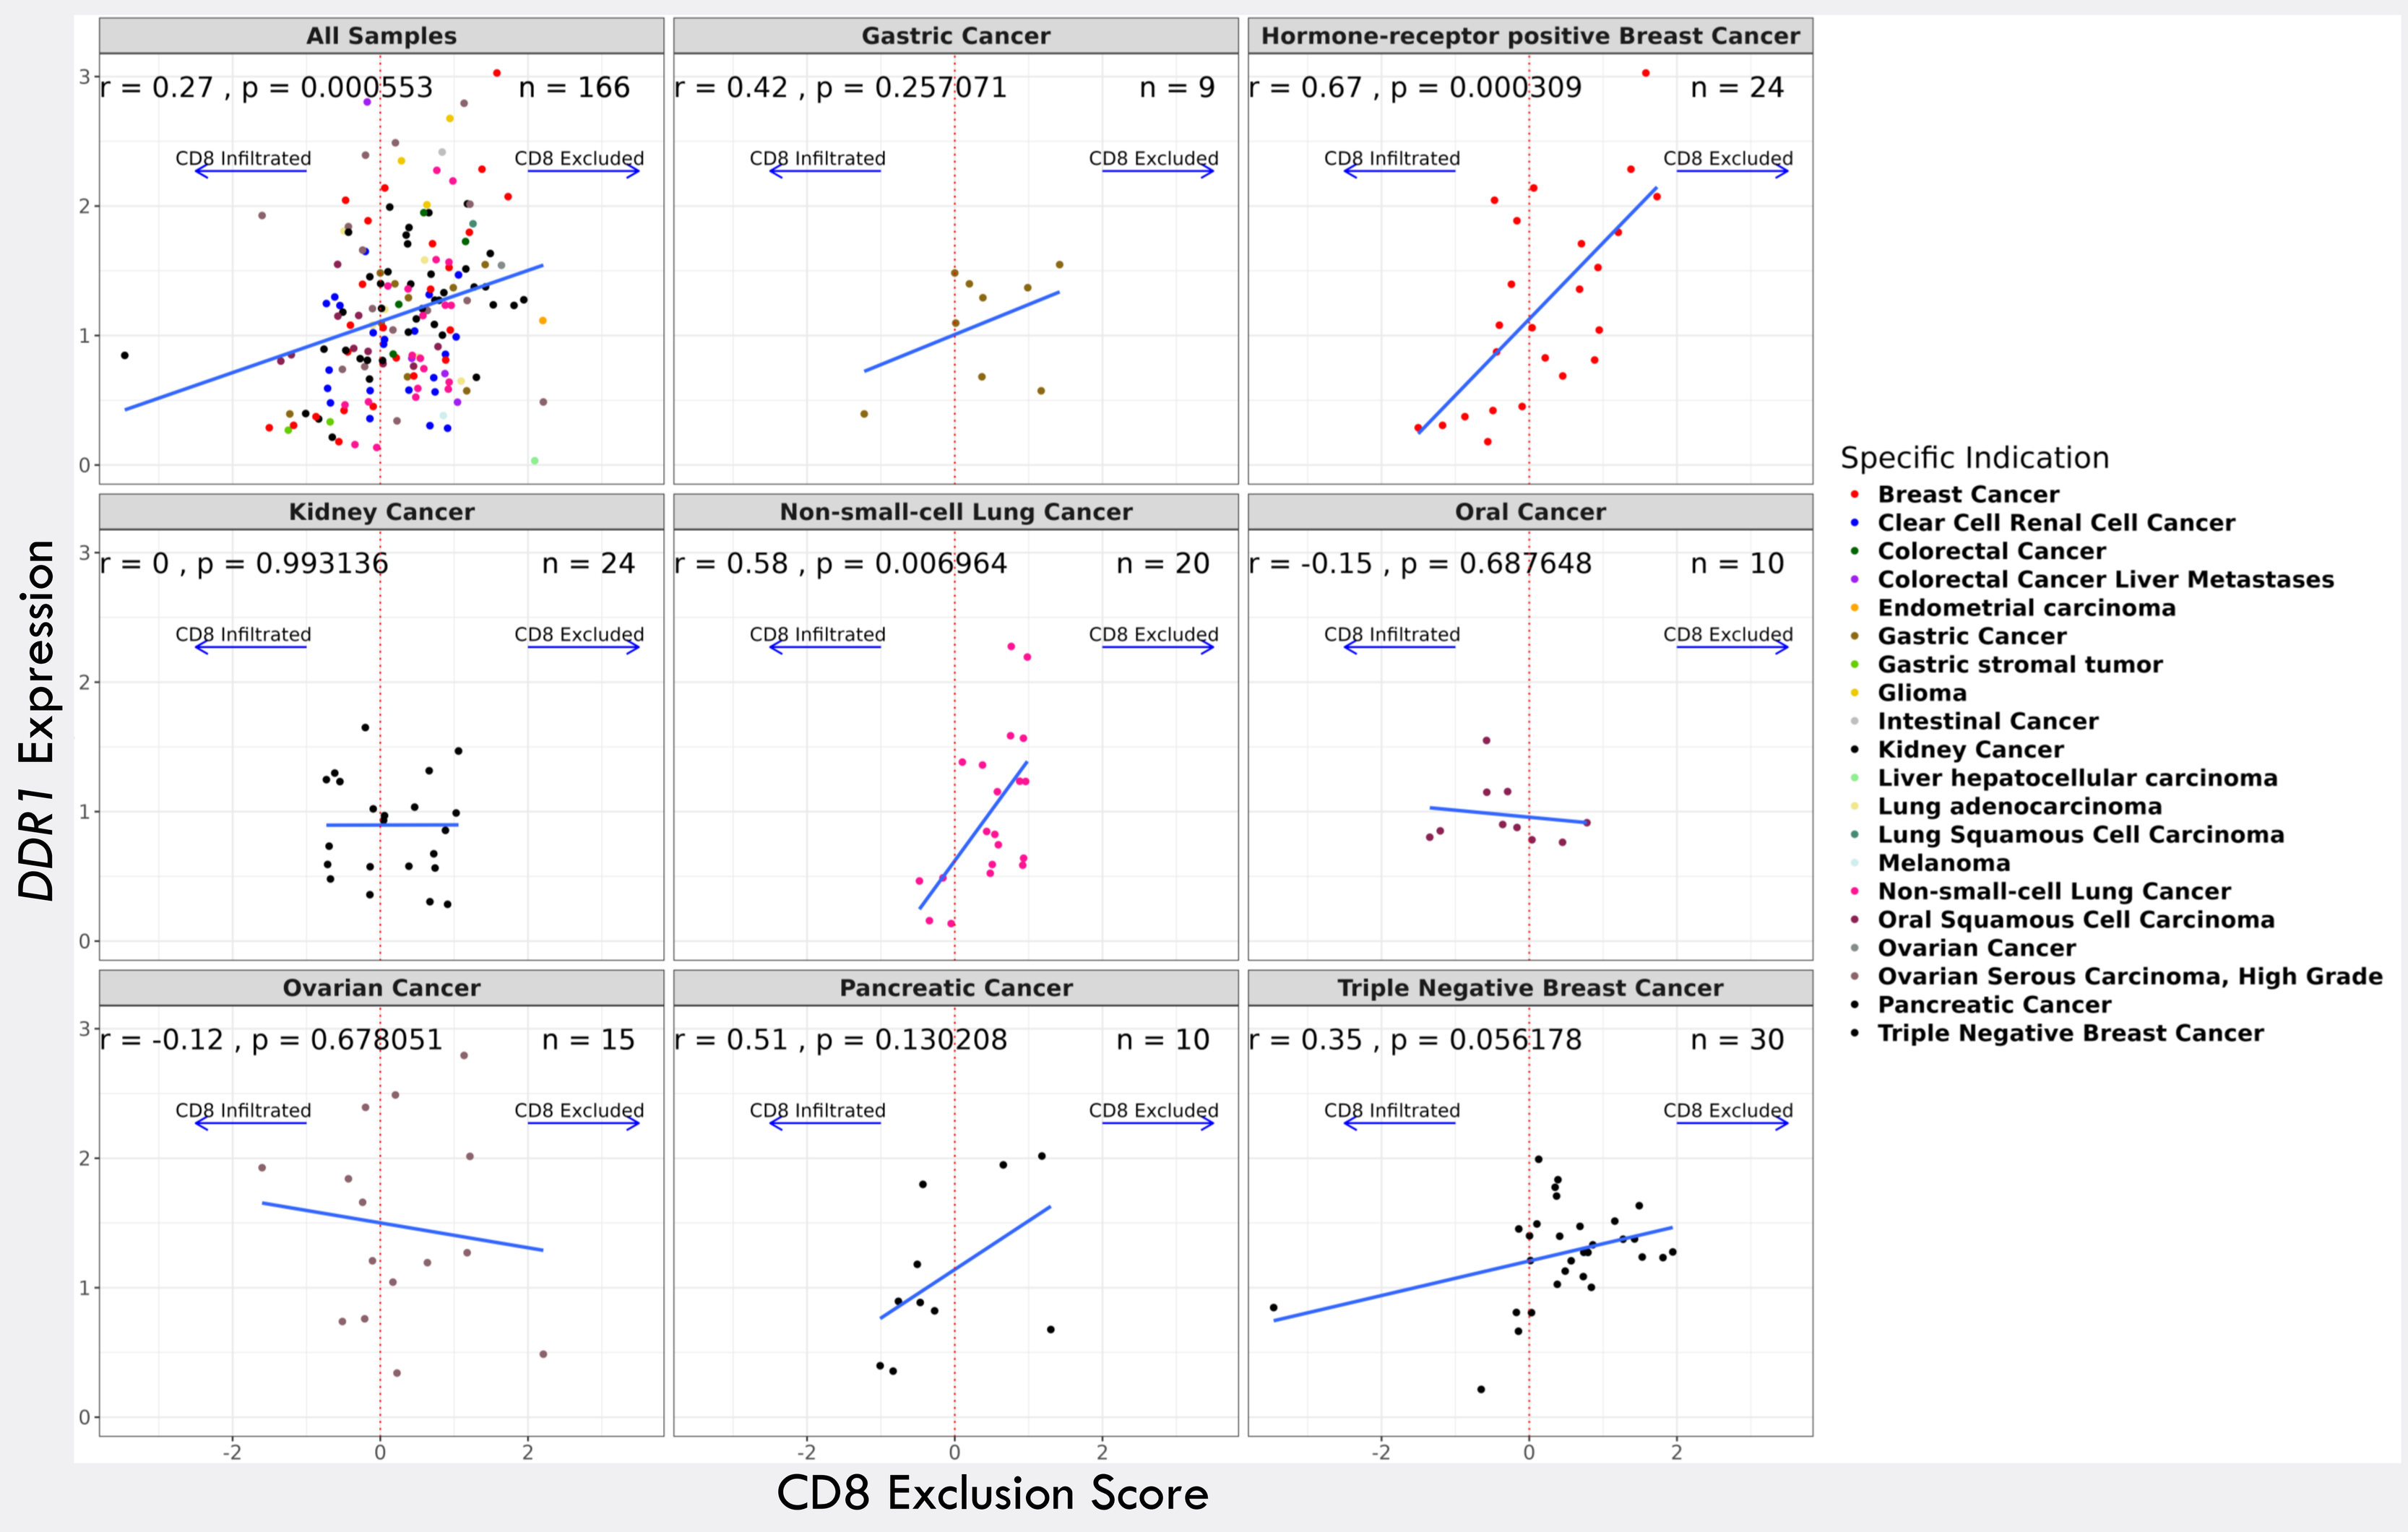
All Samples
r = 0.27 , p = 0.000553 n = 166
Gastric Cancer
r = 0.42 , p = 0.257071 n = 9
Hormone-receptor positive Breast Cancer
r = 0.67 , p = 0.000309 n = 24
Kidney Cancer
r = 0 , p = 0.993136 n = 24
Non-small-cell Lung Cancer
r = 0.58 , p = 0.006964 n = 20
Oral Cancer
r = -0.15 , p = 0.687648 n = 10
Ovarian Cancer
r = -0.12 , p = 0.678051 n = 15
Pancreatic Cancer
r = 0.51 , p = 0.130208 n = 10
Triple Negative Breast Cancer
r = 0.35 , p = 0.056178 n = 30
CD8 Infiltrated
CD8 Excluded
DDR1 Expression
CD8 Exclusion Score
Specific Indication
Breast Cancer
Clear Cell Renal Cell Cancer
Colorectal Cancer
Colorectal Cancer Liver Metastases
Endometrial carcinoma
Gastric Cancer
Gastric stromal tumor
Glioma
Intestinal Cancer
Kidney Cancer
Liver hepatocellular carcinoma
Lung adenocarcinoma
Lung Squamous Cell Carcinoma
Melanoma
Non-small-cell Lung Cancer
Oral Squamous Cell Carcinoma
Ovarian Cancer
Ovarian Serous Carcinoma, High Grade
Pancreatic Cancer
Triple Negative Breast Cancer

Figure 3: Association between *DDR1* gene expression and CD8 exclusion score. Correlation and p-values given for all samples and indications with sample size greater than or equal to 10 .

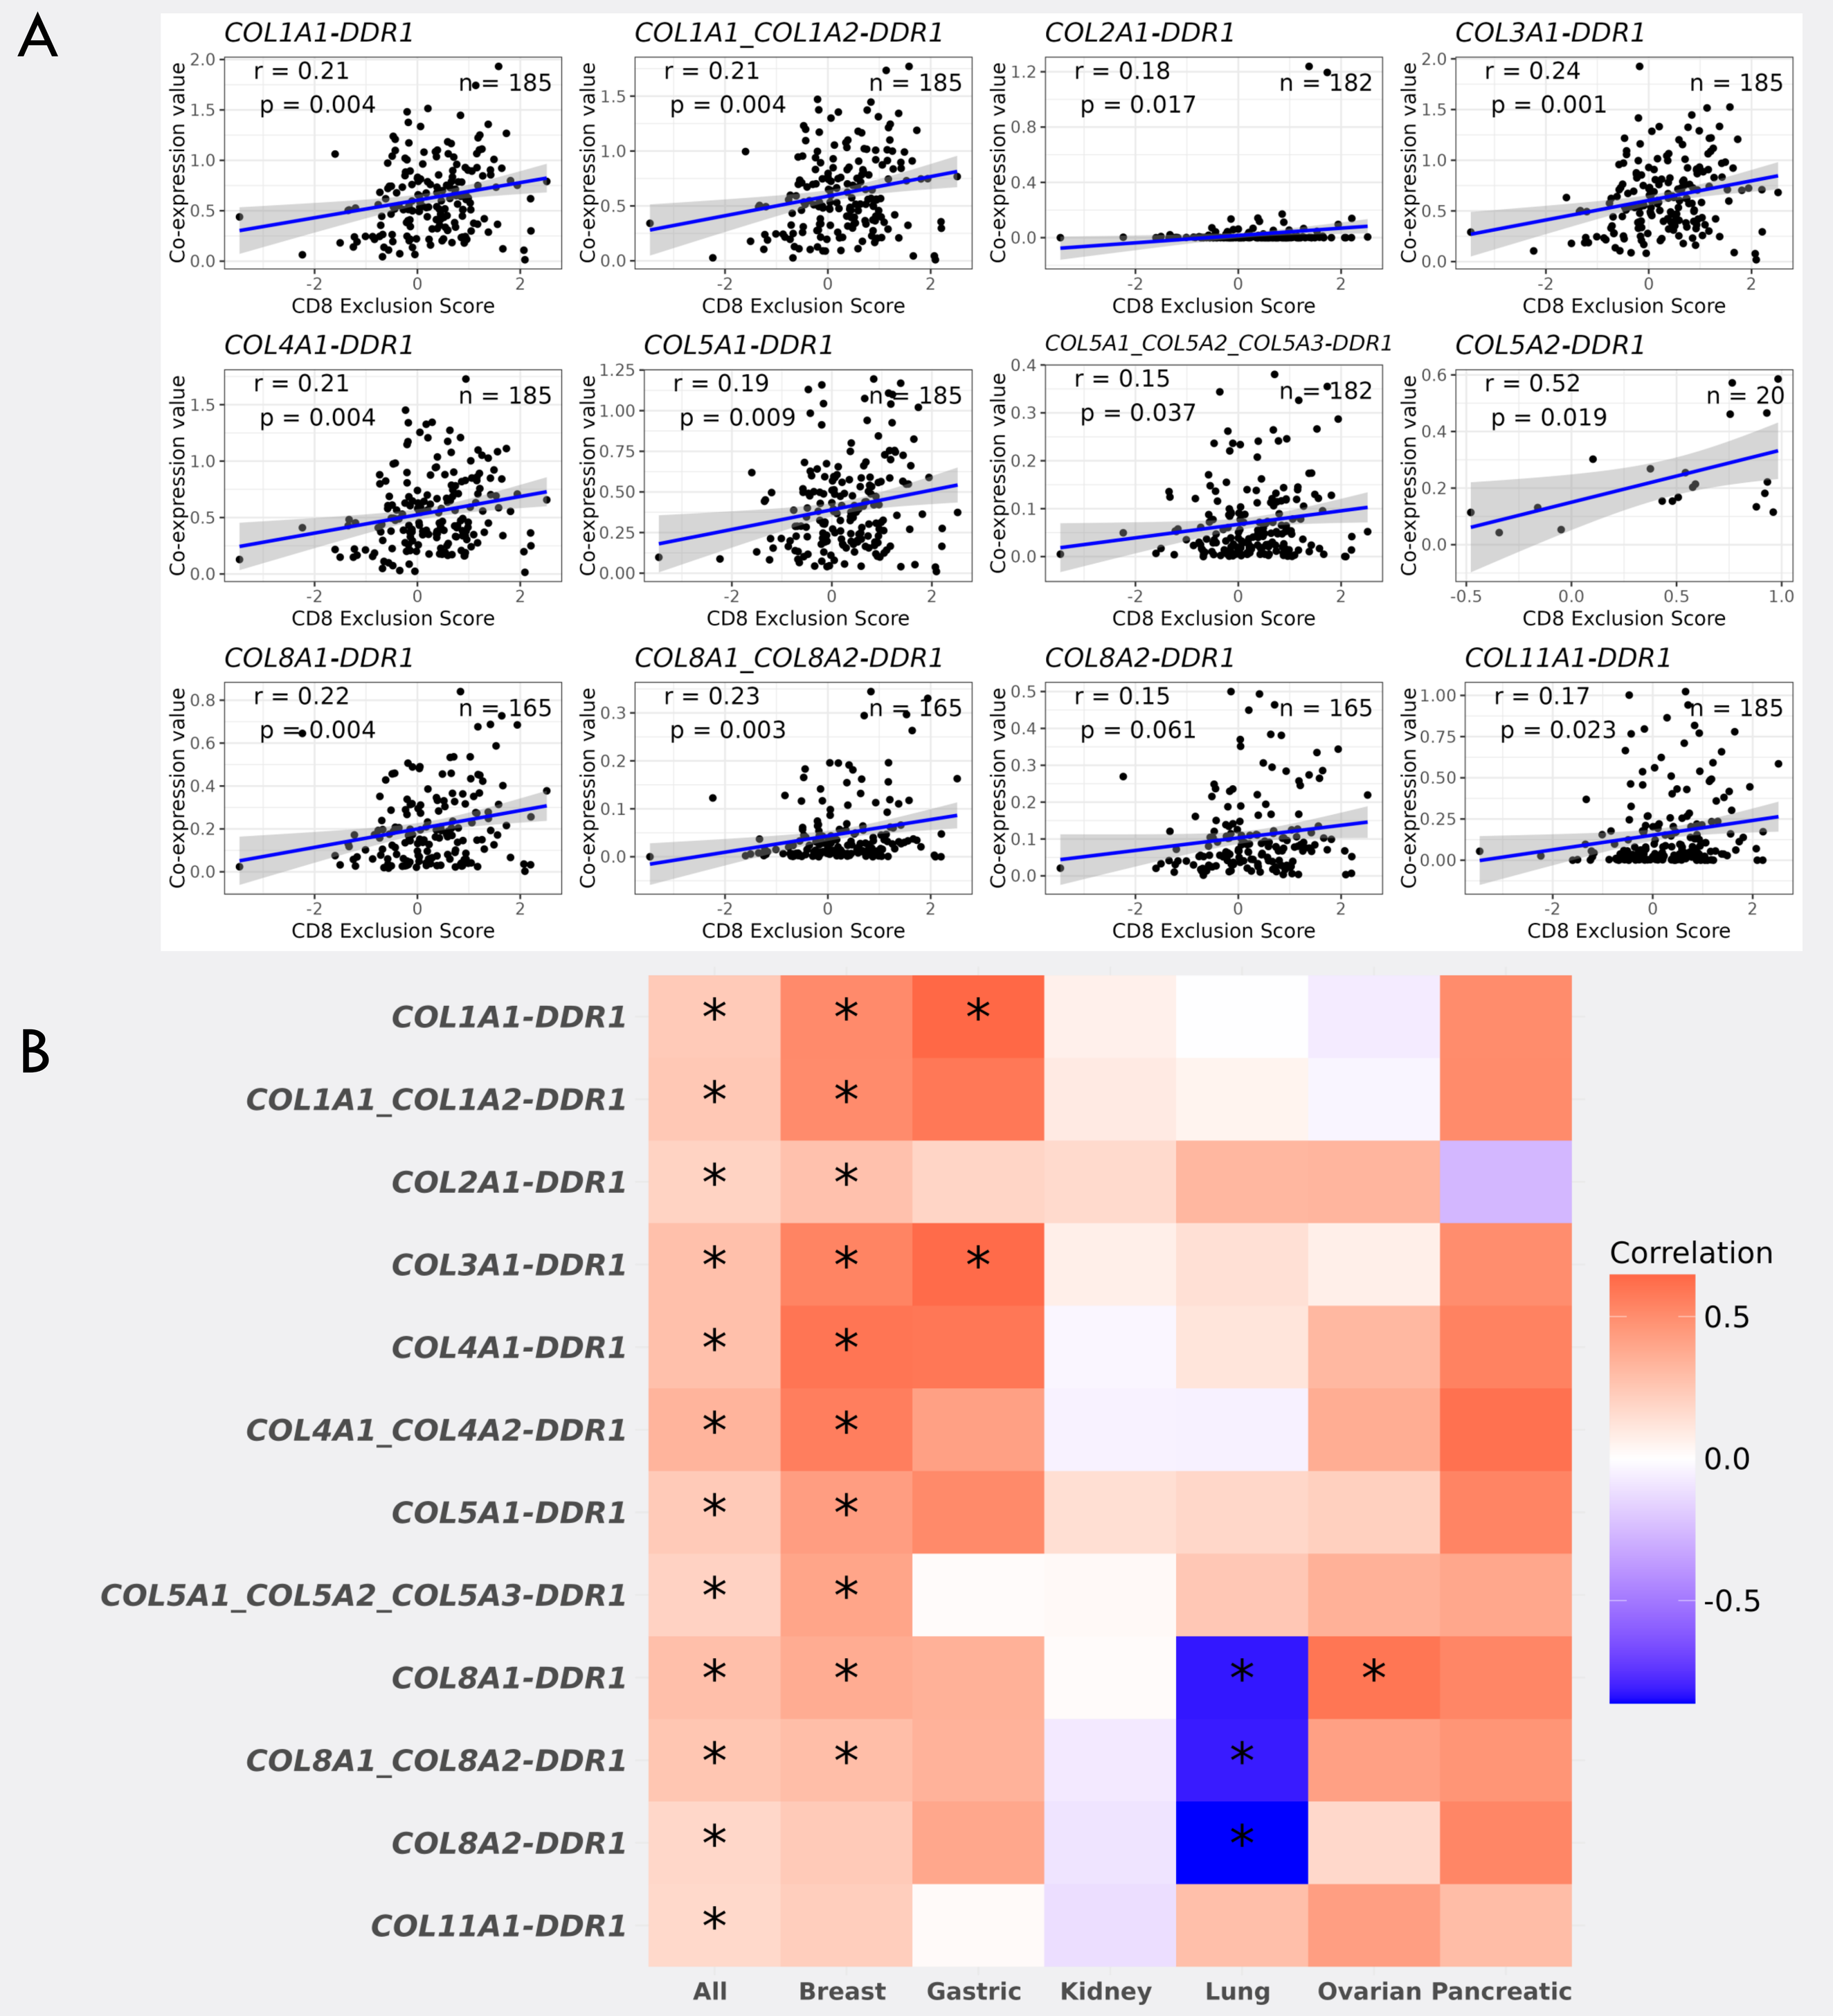
A
COL1A1-DDR1
r = 0.21 p = 0.004 n = 185
COL1A1_COL1A2-DDR1
r = 0.21 p = 0.004 n = 185
COL2A1-DDR1
r = 0.18 p = 0.017 n = 182
COL3A1-DDR1
r = 0.24 p = 0.001 n = 185
COL4A1-DDR1
r = 0.21 p = 0.004 n = 185
COL5A1-DDR1
r = 0.19 p = 0.009 n = 185
COL5A1_COL5A2_COL5A3-DDR1
r = 0.15 p = 0.037 n = 182
COL5A2-DDR1
r = 0.52 p = 0.019 n = 20
COL8A1-DDR1
r = 0.22 p = 0.004 n = 165
COL8A1_COL8A2-DDR1
r = 0.23 p = 0.003 n = 165
COL8A2-DDR1
r = 0.15 p = 0.061 n = 165
COL11A1-DDR1
r = 0.17 p = 0.023 n = 185
Co-expression value
CD8 Exclusion Score
B
COL1A1-DDR1
COL1A1_COL1A2-DDR1
COL2A1-DDR1
COL3A1-DDR1
COL4A1-DDR1
COL4A1_COL4A2-DDR1
COL5A1-DDR1
COL5A1_COL5A2_COL5A3-DDR1
COL8A1-DDR1
COL8A1_COL8A2-DDR1
COL8A2-DDR1
COL11A1-DDR1
All
Breast
Gastric
Kidney
Lung
Ovarian
Pancreatic
Correlation
0.5
0.0
-0.5

Figure 4. Association between *DDR1*-collagen co-expression and CD8 exclusion score. A). Scatterplots for all *DDR1* and collagen co-expression versus immune exclusion score shown with correlation and p-values. B) Heatmap of correlation between *DDR1* and collagen co-expression with CD8 exclusion score. Red represents positive correlation, and blue represents negative correlation. * signifies statistical significance (p-value < 0.05).

A

Rank	Pathway
1	EPHA
2	WNT
3	CDH
4	GDF
5	ncWNT
6	DDR1/2:Collagen
7	EPHB
8	PSAP
9	HSPG
10	GRN
11	BMP
12	LAMININ
13	AGRN
14	BSP
15	DESMOSOME
16	ENHO
17	MK
18	ACTIVIN
19	JAM
20	OCLN
21	COLLAGEN
22	GALANIN
23	NOTCH
24	CD46
25	SPP1

B

Enrichment of DDR1-Collagen interactions in the ranked list of LR interactions enriched in CD8 excluded samples.

Core Enrichment: COL3A1-DDR1, COL1A1_COL1A2-DDR1, COL5A2-DDR1, COL1A1-DDR1, COL4A1-DDR1, COL4A1_COL4A2_COL4A3_COL4A4-DDR1

NES: 1.85, p-value: 0.00077

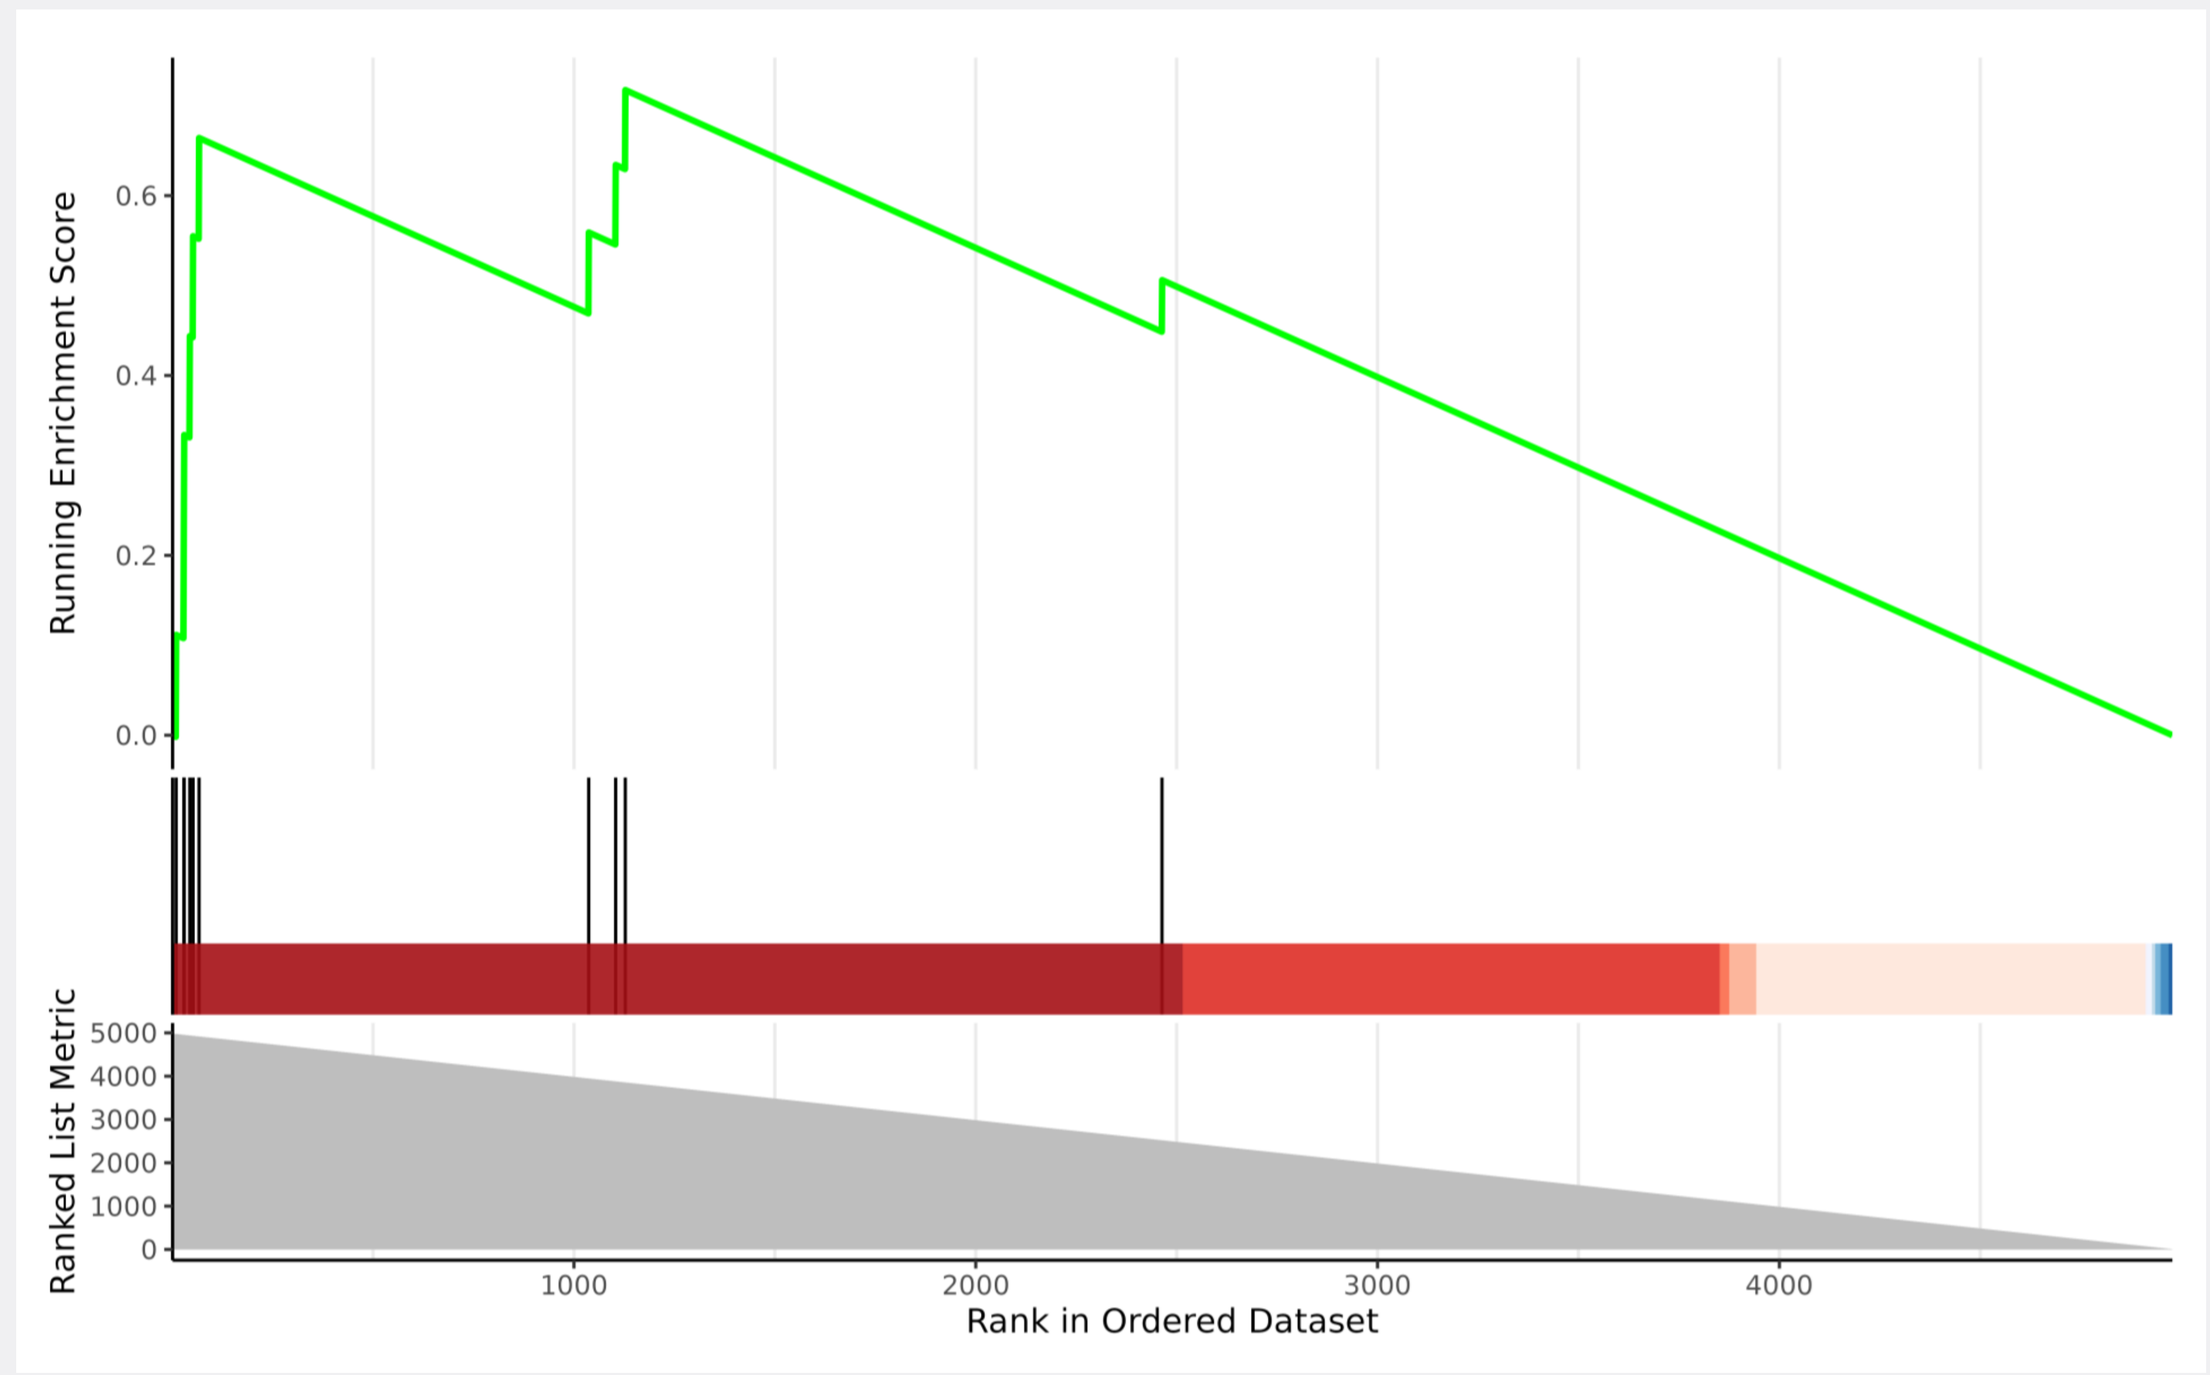
Running Enrichment Score
Ranked List Metric
Rank in Ordered Dataset

Figure 5. Top Ranked Ligand-Receptor Interactions and Pathways Associated with Immune Exclusion
A) Top 25 Pathways Associated with Immune Exclusion Based on Differential Co-Expression Analysis.
B) Hypergeometric Plot Showing the Enrichment of DDR1 and Collagen Interactions in the Ranked List of Immune Excluded Ligand-Receptor Interactions.

Conclusions

We developed a pipeline to quantify spatially-resolved gene expression across various human tumors. *DDR1* mRNA and *DDR1*-collagen co-expression are correlated with the degree of CD8 exclusion with varying strength of associations across tumor types. These insights are valuable for better understanding the biology of DDR1 in the tumor microenvironment.

References

1. Sun, X et al. *Nature*. 2021.
2. Sher, X et al. *Cancer Res*. 2024.
3. Arora, R et al. *Nat Commun.* 2023.
4. Barkley, D et al. *Nat Genet*. 2022.
5. Bassiouni, R et al. *Cancer Res*. 2023.
6. Coutant, A et al. *Lab Invest*. 2023.
7. Denisenko, E et al. *Nat Commun*. 2024.
8. De Zuani, M et al. *BioStudies*. 2024.
9. Garbarino, O et al. *Aging Cell*. 2023.
10. Human Lung Cancer, 11 mm Capture Area (FFPE), 10x Genomics.
11. Jang, Ei et al. *Exp Mol Med*. 2023.
12. Kim, S et al. *Genome Med*. 2024.
13. Lyubetskaya, A et al. *Cell Rep Methods*. 2022.
14. Mei, Y et al. *Nat Cancer*. 2023.
15. Meylan, M et al. *Immunity*. 2022.
16. Ren, Y et al. *Nat Commun*. 2023.
17. Stur, E, et al. *iScience*. 2022.
18. Tashireva, L et al. *Discov Med*. 2023.
19. Various Tumors, Spatial Gene Expression by Space Ranger, 10x Genomics.
20. Villemin, J et al. *Nucleic Acids Res*. 2023.
21. Zhu, J et al. *Exp Mol Med*. 2022.
22. Shih, AJ et al. *PLoS one*. 2018.
23. Jerby-Arnon, L et al. *Cell*. 2018.
24. Cang, Z et al. *Nat. methods*. 2023.