Stromal collagen features from H&E-stained whole slide images are associated with lymphocyte infiltration and survival following checkpoint inhibition in patients with non-small cell lung cancer

G. Travis Clifton[1*^], Ciyue Shen[2*], Nhat Le[2], Jun Zhang[2], Jennifer Hipp[2], Tan Nguyen[2], Bahar Rahsepar[2], Michael Griffin[2], Jacqueline Brosnan-Cashman[2], Justin Lee[2#], Robert Egger[2], Jean-Philippe Guégan[3], Alban Bessede[3], Florent Peyraud[4], Antoine Italiano[4], Xinwei Sher[1], Matthew Bronnimann[2*], Laura A. Dillon[1*^]

[1]Incendia Therapeutics, Cambridge, MA; [2]PathAI, Boston, MA; [3]Explicyte, Bordeaux, France; [4]Institut Bergonie, Bordeaux, France; ^Employee of Incendia Therapeutics at the time of study; #Employee of PathAI at the time of study; *Contributed equally to study

Abstract #1378

STUDY BACKGROUND

- Immunotherapy, especially immune checkpoint inhibition (CPI), has become a common treatment strategy for NSCLC. Many agents targeting PD-1 (e.g., nivolumab and pembrolizumab) and PD-L1 (e.g., atezolizumab) have been granted FDA approval for the treatment of NSCLC in patients expressing PD-L1 [1-3].
- Extracellular matrix components, including stromal collagen, may prevent infiltration of tumors by T cells, resulting in poor CPI response to CPI [4].
- To assess how collagen structure may influence clinical outcomes following CPI therapy, we developed an approach to extract and compute collagen fiber-based features from hematoxylin and eosin (H&E)-stained tumor sections, which we associate with CPI outcomes and lymphocyte infiltration in a NSCLC cohort.

DETECTION OF COLLAGEN AND TME FEATURES FROM H&E

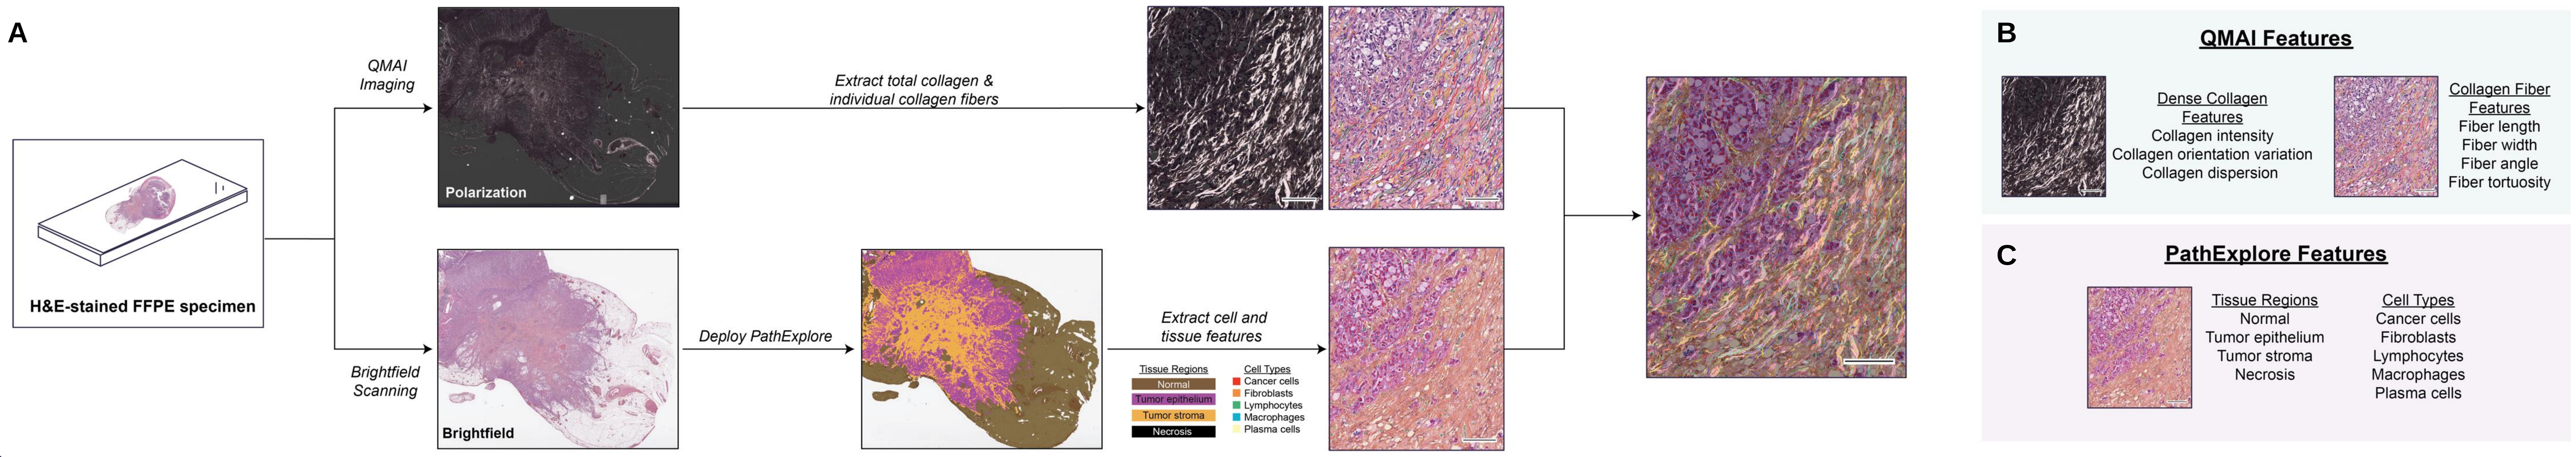
A
H&E-stained FFPE specimen
QMAI Imaging
Polarization
Brightfield Scanning
Brightfield
Extract total collagen & individual collagen fibers
Deploy PathExplore
Extract cell and tissue features
Tissue Regions
Normal
Tumor epithelium
Tumor stroma
Necrosis
Cell Types
Cancer cells
Fibroblasts
Lymphocytes
Macrophages
Plasma cells
B
QMAI Features
Dense Collagen Features
Collagen intensity
Collagen orientation variation
Collagen dispersion
Collagen Fiber Features
Fiber length
Fiber width
Fiber angle
Fiber tortuosity
C
PathExplore Features
Tissue Regions
Normal
Tumor epithelium
Tumor stroma
Necrosis
Cell Types
Cancer cells
Fibroblasts
Lymphocytes
Macrophages
Plasma cells

Figure 1. Integration of QMAI and PathExplore for examining the interplay between collagen and the tumor microenvironment. A) Study overview. The merged overlay shows QMAI and PathExplore outputs. Scale bars indicate 100 µm. Features are extracted and computed by B) QMAI and C) PathExplore.

METHODS

Dataset

- PD-(L)1 inhibitor-treated NSCLC patients (N=95) were enrolled in the Bergonié Institut Profiling precision medicine study (NCT02534649; Institut Bergonié, Bordeaux, France). Pre-treatment specimens, stained with H&E, were imaged with a Leica AT2 scanner at 40X magnification. Clinical metadata included disease histology, PD-L1 tumor proportion score (TPS), and clinical outcome data for each patient.

QMAI and Collagen Feature Extraction

- The imaging workflow is shown in Fig. 1A. Quantitative Multimodal Anisotropy Imaging (QMAI), a polarization-based modality to image tissue components with birefringence (e.g., collagen fibers), was performed using a custom Olympus BX63 microscope. Polarized and brightfield whole-slide images (WSIs) were captured. Brightfield WSIs were used to match WSIs at pixel-level accuracy, and polarization WSIs were used for fiber extraction, which was performed as described [5].
- Dense collagen features computed were collagen intensity, orientation variation, and dispersion; collagen fiber features features computed were fiber length, width, relative angle, and tortuosity (Fig. 1B).

Tissue and Cell Classification and Feature Extraction

- NSCLC-specific PathExplore™ models (PathAI, Boston, MA) were used to predict tissue regions [6] and cell types [6,7] in H&E-stained WSIs.
- Cell models predicted cancer epithelial cells, fibroblasts, macrophages, lymphocytes, and plasma cells, and tissue models predicted normal tissue, tumor epithelium, tumor stroma, and necrosis (Fig. 1C). Regions 60, 120, and 120+ µm from the epithelial-stromal interface (ESI; Fig. 2) were also measured. Regions predicted to be tumor stroma were used for collagen feature extraction.
- Immune phenotypes (IPs) were predicted using a patch-based approach incorporating PathExplore-derived lymphocyte density measurements, as previously described [8].

Figure 2. Epithelial-stromal interface measurement in NSCLC.

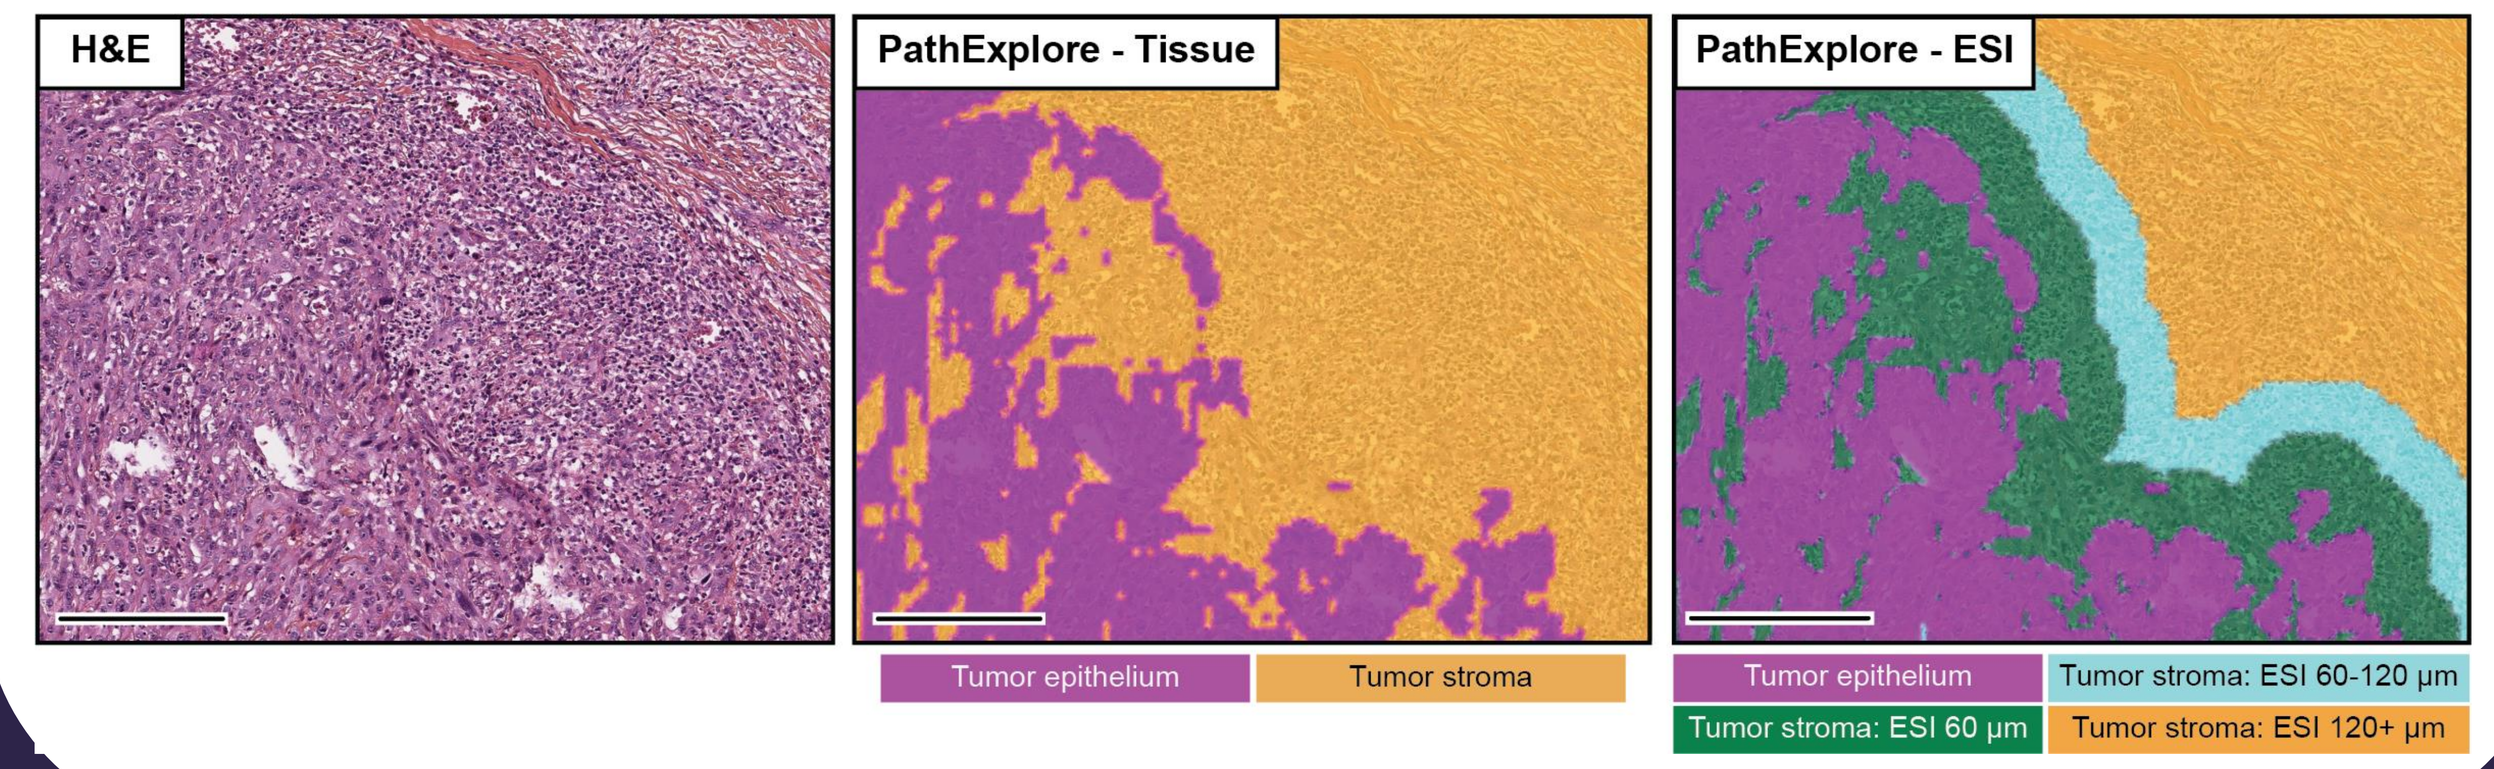
H&E
PathExplore - Tissue
PathExplore - ESI
Tumor epithelium
Tumor stroma
Tumor epithelium
Tumor stroma: ESI 60-120 µm
Tumor stroma: ESI 60 µm
Tumor stroma: ESI 120+ µm

RESULTS

- In regions close to the ESI, fibers are narrower (Fig. 3A), less tortuous (Fig. 3B), and oriented more parallel to the ESI (Fig. 3C) compared to regions distal to the ESI. In cases with PD-L1 TPS <1%, median collagen intensity in regions 60-120 µm from the ESI (Fig. 4A), and fiber width and tortuosity in stroma (Fig. 4B,C) were significantly greater in total tumor stroma compared to tumors with PD-L1 TPS >1%.
- Median collagen intensity in regions 60-120 µm from the ESI and fiber width and tortuosity in tumor stroma were significantly lower in cases with partial or complete response to CPI, compared to those with stable or progressive disease (Fig. 5A). When PD-L1 status and histologic subtypes were also considered, fiber width, and to some extent fiber tortuosity, were also correlated with response to CPI (Fig. 5B). Furthermore, there was better overall survival (OS) in patients with thinner stromal collagen fibers, in both PD-L1 low and high groups, especially in patients with high PD-L1 TPS (Fig. 5C).
- Lymphocyte density was significantly lower with greater fiber width and tortuosity (Fig. 6A) in both tumor epithelium and stroma. Both fiber width and tortuosity (Fig. 6B) were greater in tumors predicted to have an immune desert phenotype, compared to tumors predicted to have an inflamed phenotype.

Figure 3. Changes in collagen features with increasing distance from the tumor boundary.

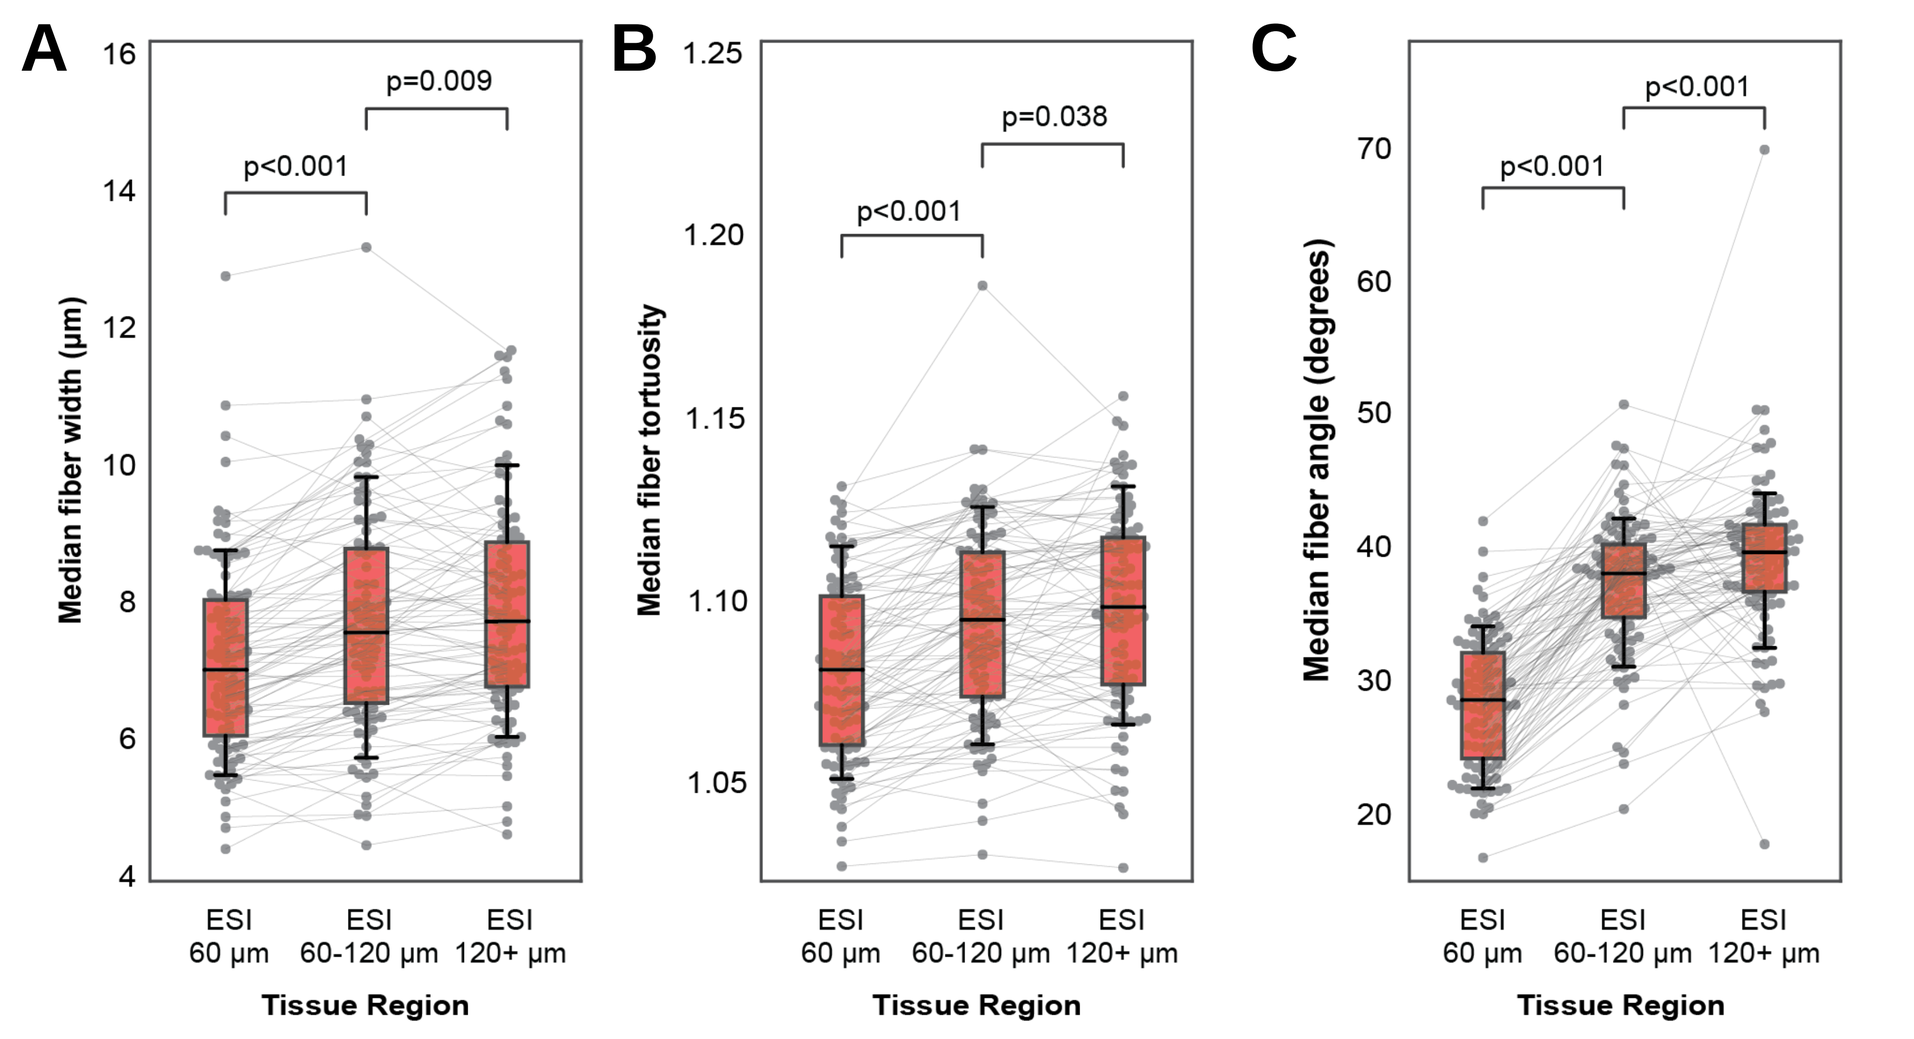
A
p<0.001
p=0.009
Median fiber width (µm)
ESI 60 µm
ESI 60-120 µm
ESI 120+ µm
Tissue Region
B
p<0.001
p=0.038
Median fiber tortuosity
C
p<0.001
p<0.001
Median fiber angle (degrees)

Changes in QMAI-derived A) fiber width, B) fiber tortuosity, and C) fiber angle are shown as the distance from the ESI increases. Median values of these features are shown.

Figure 4. Association of collagen features with PD-L1 status.

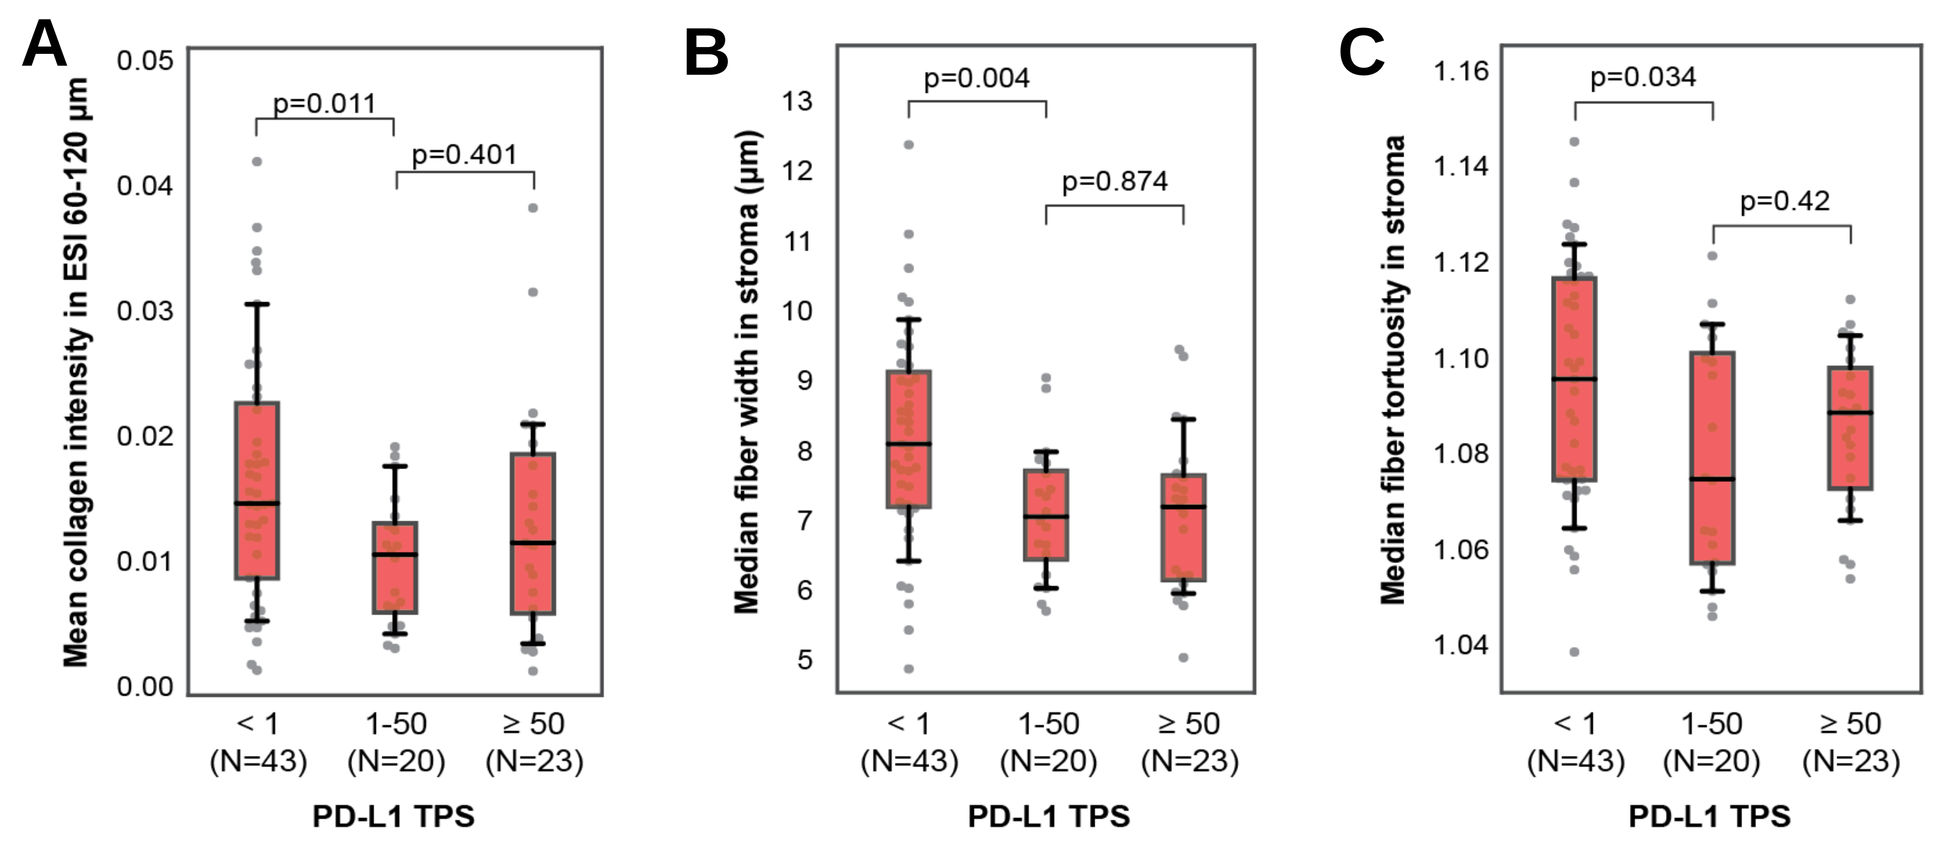
A
p=0.011
p=0.401
Mean collagen intensity in ESI 60-120 µm
< 1 (N=43)
1-50 (N=20)
≥ 50 (N=23)
PD-L1 TPS
B
p=0.004
p=0.874
Median fiber width in stroma (µm)
C
p=0.034
p=0.42
Median fiber tortuosity in stroma

QMAI-derived collagen intensity in ESI 60-120 µm (A), fiber width (B) and fiber tortuosity (C) were compared in NSCLC tumors with varying PD-L1 TPS.

Figure 5. Association between collagen fiber features and response to CPI in NSCLC.

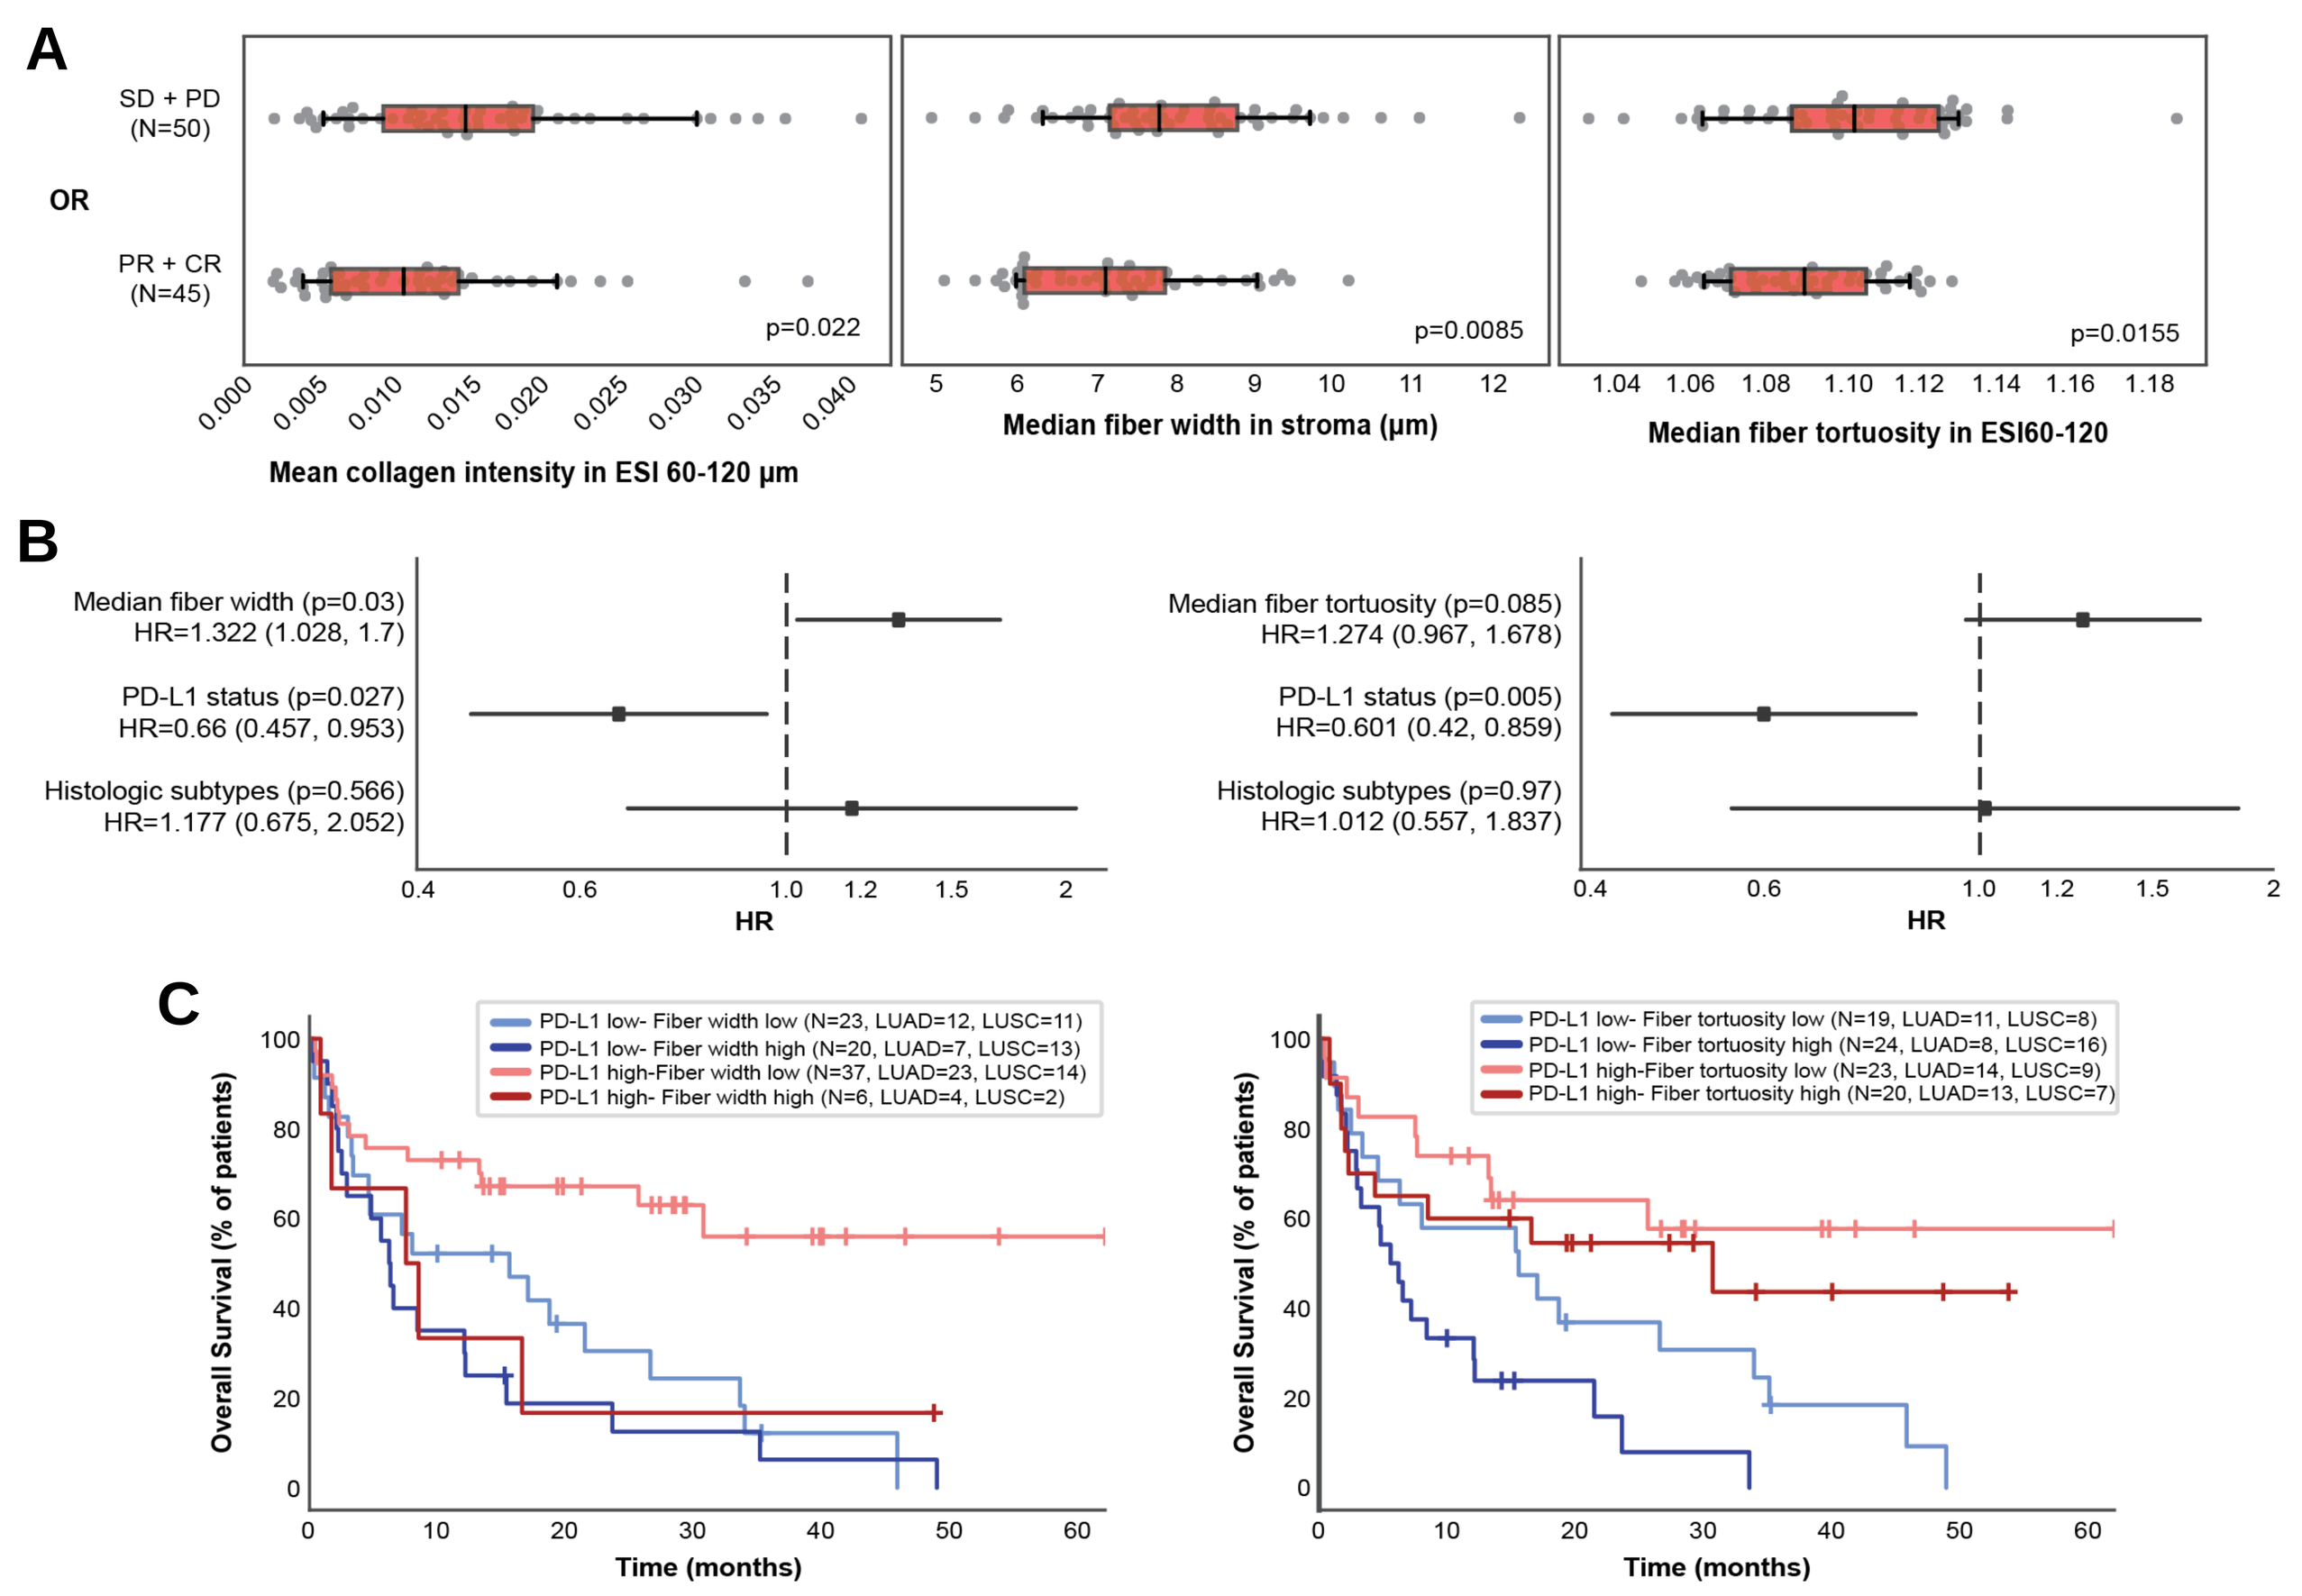
A
SD + PD (N=50)
OR
PR + CR (N=45)
p=0.022
p=0.0085
p=0.0155
Mean collagen intensity in ESI 60-120 µm
Median fiber width in stroma (µm)
Median fiber tortuosity in ESI60-120
B
Median fiber width (p=0.03) HR=1.322 (1.028, 1.7)
PD-L1 status (p=0.027) HR=0.66 (0.457, 0.953)
Histologic subtypes (p=0.566) HR=1.177 (0.675, 2.052)
Median fiber tortuosity (p=0.085) HR=1.274 (0.967, 1.678)
PD-L1 status (p=0.005) HR=0.601 (0.42, 0.859)
Histologic subtypes (p=0.97) HR=1.012 (0.557, 1.837)
HR
C
PD-L1 low- Fiber width low (N=23, LUAD=12, LUSC=11)
PD-L1 low- Fiber width high (N=20, LUAD=7, LUSC=13)
PD-L1 high-Fiber width low (N=37, LUAD=23, LUSC=14)
PD-L1 high- Fiber width high (N=6, LUAD=4, LUSC=2)
PD-L1 low- Fiber tortuosity low (N=19, LUAD=11, LUSC=8)
PD-L1 low- Fiber tortuosity high (N=24, LUAD=8, LUSC=16)
PD-L1 high-Fiber tortuosity low (N=23, LUAD=14, LUSC=9)
PD-L1 high- Fiber tortuosity high (N=20, LUAD=13, LUSC=7)
Overall Survival (% of patients)
Time (months)

A) Collagen intensity (in ESI 60-120 µm) and fiber width and tortuosity were compared in patients classified as non-responders (SD+PD) and responders (PR+CR). B) Forest plots showing the association of QMAI-derived fiber width and fiber tortuosity to overall survival, considering PD-L1 TPS and histologic subtype as additional covariates. C) Kaplan-Meier curves stratifying patients by PD-L1 (TPS ≥ 1%) and QMAI-derived fiber width or tortuosity to predict OS.

Figure 6. Association between collagen fiber features and immune phenotypes.

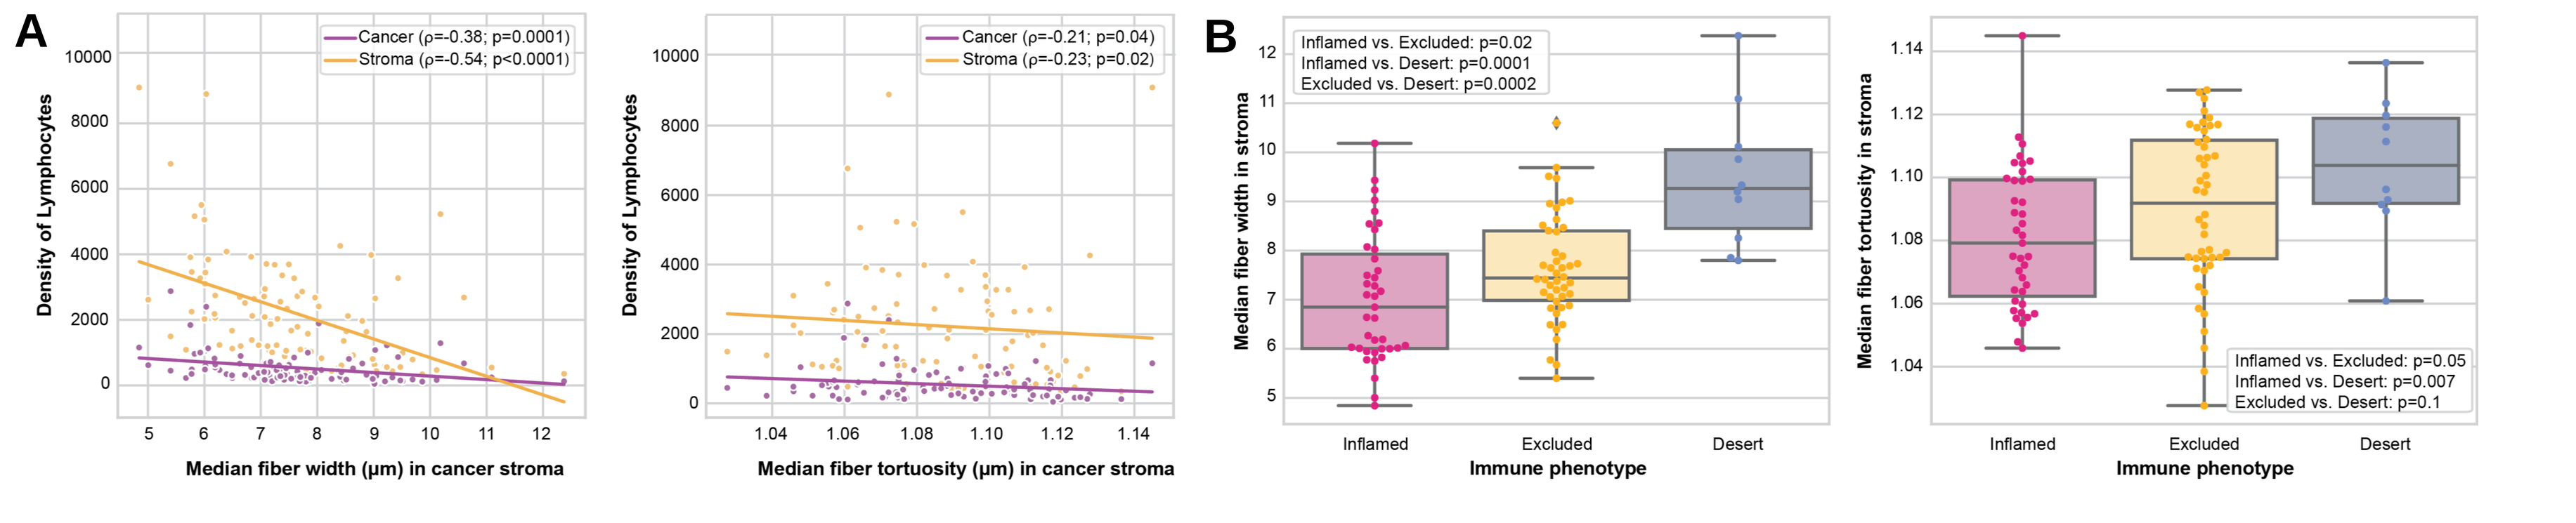
A
Cancer (ρ=-0.38; p=0.0001)
Stroma (ρ=-0.54; p<0.0001)
Density of Lymphocytes
Median fiber width (µm) in cancer stroma
Cancer (ρ=-0.21; p=0.04)
Stroma (ρ=-0.23; p=0.02)
Median fiber tortuosity (µm) in cancer stroma
B
Inflamed vs. Excluded: p=0.02
Inflamed vs. Desert: p=0.0001
Excluded vs. Desert: p=0.0002
Median fiber width in stroma
Inflamed
Excluded
Desert
Immune phenotype
Median fiber tortuosity in stroma
Inflamed vs. Excluded: p=0.05
Inflamed vs. Desert: p=0.007
Excluded vs. Desert: p=0.1

A) Correlation of median QMAI-derived fiber width and tortuosity with lymphocyte density using Spearman rank correlation analysis. B) Median QMAI-derived fiber width and tortuosity in stroma in tumors predicted to be inflamed, excluded, or desert. Significance was calculated using Mann Whitney U test.

CONCLUSIONS

- Using QMAI, an imaging modality that extracts features of individual collagen fibers from H&E-stained WSIs, we show that increased collagen fiber width is associated with low lymphocyte infiltration, low PD-L1 levels, and non-response in CPI-treated NSCLC.
- These results support the hypothesis that excessive collagen promotes immune exclusion by blocking lymphocytes from entering the tumor epithelium, supporting the development of therapies to alter collagen alignment (Fig. 7).

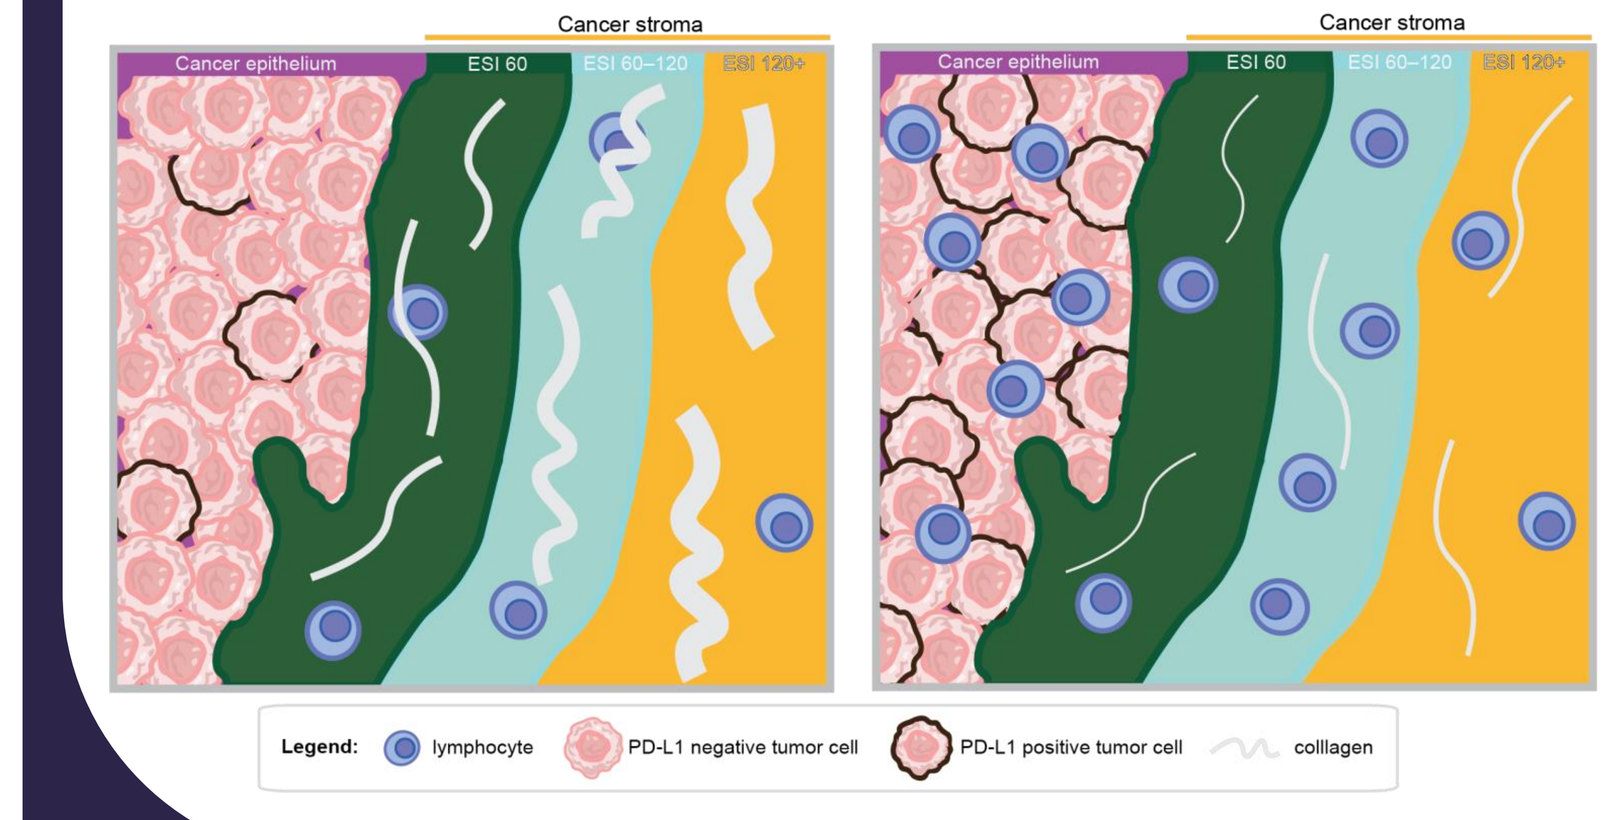
Cancer stroma
Cancer epithelium
ESI 60
ESI 60–120
ESI 120+
Legend:
lymphocyte
PD-L1 negative tumor cell
PD-L1 positive tumor cell
collagen

Figure 7. Hypothesis. Thick, tortuous collagen bundles (left) prevent infiltration of lymphocytes into the tumor epithelium. Thin, straight fiber bundles (right) allow infiltration.

REFERENCES

1) Kazandjian, D., et al. *Oncologist*. 2016; 21:6344-42.
2) Sul, J., et al., Oncologist. 2016;21:643–50.
3) Weinstock, C., et al. Clin Cancer Res. 2017;23:4534–9.
4) Peng, D.H., et al. *Nat Commun*. 2020; 11:4520.
5) Bredfeldt, J.S., et al. *J Biomed Opt*. 2014; 19:016007.
6) Markey, M. and Kim, J. et al., *bioRxiv* 2024.08.12.607604.
7) Abel, J., et al. *npi Precis Onc*. 2024; 8:134.
8) Le, N. et al., *J Clin Oncol.* 2024; 42, 8539.

* PathExplore is for research use only. Not for use in diagnostic procedures.

ACKNOWLEDGMENTS

We would like to thank the patients involved in this study and their families, as well as members of the PathAI Pathology Network who contributed towards this work. We also thank Lara Murray (PathAI) for assistance with figure design.

This poster template was developed by SciStories LLC. https://scistories.com/

CONTACT: matthew.bronnimann@pathai.com

Society for Immunotherapy of Cancer 39th Annual Meeting November 6-10, 2024, Houston, TX